A1-2015

SIMILARITY IN PHARMACOKINETICS OF TBR-652, A CHEMOKINE RECEPTOR 5 (CCR5) ANTAGONIST, IN HEALTHY AND HIV-1 INFECTED SUBJECTS

David E. Martin[1], Leng Hong Pheng[2], My My Trinh[2], JF Marier[2], Reynold Driz[1], Richard Ogden[1], Sandra M. Palleja[1] • [1]Tobira Therapeutics, Inc., Manalapan, NJ, USA; [2]Pharsight, a Certara Company, Montreal, Quebec, Canada

David E. Martin, PharmD, MBA
198 US Highway 9 North, Suite 200
Manalapan, New Jersey 07726
(732) 863-5980
dmartin@tobiratherapeutics.com

INTRODUCTION

TBR-652 is a potent CCR5 antagonist currently in phase 2 development for the treatment of HIV-1 infection. In a recently completed 10-day monotherapy study, median nadir reductions in plasma HIV-1 RNA levels of up to 1.8 $\log_{10}$ copies/mL were observed in antiretroviral experienced patients with CCR5-tropic virus.[1] The pharmacokinetics (PK) of TBR-652 on Days 1 and/or 10 after once-daily (QD) administration of ascending doses of TBR-652 for 10 days have been investigated in healthy subjects under fasted and fed conditions.[2,3] The PK of TBR-652 have also been reported in HIV-infected patients administered ascending doses of TBR-652 for 10 days.[4]

OBJECTIVES

To determine the PK of TBR-652 following single and repeated oral administrations under fasted and fed conditions in healthy and HIV-1 infected subjects.

METHODS

PK Studies in Healthy Subjects

Study 652-1-101 was a single-dose, double-blind, randomized, placebo-controlled study, at doses ranging from 10 mg to 800 mg in 24 healthy subjects under fasted conditions. Study 652-1-102 was a two-part, crossover study in 96 healthy subjects. In Part 1, three groups of subjects under fasted and fed (high- and low-fat breakfast) conditions received an open-label, single 100 mg TBR-652 tablet sequentially. Part 2 of study 652-1-102 was a double-blind, placebo-controlled, randomized design evaluating multiple doses of oral TBR-652 administered QD for 10 days at doses ranging from 25 to 200 mg following a high-fat breakfast. All studies explored two formulations: 25 mg tablets in the original formulation (F1) and 100 mg tablets in newer formulation (F2).

PK Study in HIV-1 Infected Subjects

Study 652-2-201 was a double-blind, placebo-controlled, randomized study of oral TBR-652 QD for 10 days, in a total of 54 HIV-1 infected patients at doses of 25 to 150 mg under fed conditions (low-fat breakfast). Patients were allowed to follow their normal eating habits; excessive amounts of fat were to be avoided at breakfast. Of the 54 subjects enrolled in the study, a total of 40 patients provided sufficient data for PK analysis. This study explored the 25 mg tablet in formulation F1 and the 100 mg tablet in formulation F2.

Analyses

PK parameters of TBR-652 for the 25 and 100 mg doses in healthy subjects were compared to those in HIV-1 infected subjects based on descriptive statistics. A non-compartmental PK analysis was performed on the concentration-time profiles of TBR-652 to calculate $AUC_{0\text{-}24}$, T_{max}, C_{max}, C_{min}, $t_{½}$ and CL/F following single and/or multiple QD doses of TBR-652.

RESULTS

Demographics/Disposition

A total of 55 subjects across 3 studies provided sufficient PK samples to assess individual noncompartmental PK parameters of TBR-652 in healthy and HIV-1 infected subjects (**Table 1**).

Table 1. Healthy and HIV-1 Infected Subject Demographics

		1 x 25 mg F1 Tablet			1 x 100 mg F2 Tablet		
		Healthy		HIV-1	Healthy		HIV-1
Characteristics		652-1-101 (Fasted) N=9	652-1-102 (High Fat) N=9	652-2-201 (Low Fat) N=8	652-1-102 (Low Fat) N=12	652-1-102 (High Fat) N=9	652-2-201 (Low Fat) N=8
Age (years)	Mean	25.3	42.9	41.0	46.9	34.4	39.9
	SD	4.03	7.36	7.95	6.07	8.83	6.92
	Range	21–33	31–50	24–47	36–55	19–45	29–48
	Median	25	45	44	47	39	39
Gender (n, %)	Male	6 (66.7)	3 (33)	8 (100.0)	5 (42)	4 (44)	8 (100.0)
	Female	3 (33.0)	6 (67)	0 (0.0)	7 (58)	5 (56)	0 (0.0)
Race (n, %)	Caucasian	1 (11.1)	8 (89)	7 (87.5)	10 (83)	8 (89)	4 (50.0)
	African American	7 (77.8)	-	-	2 (17)	-	4 (50.0)
	Asian	-	-	-	-	-	-
	Hispanic	-	1 (11)	1 (12.5)	-		-
	Other	1 (11.1)	-	-	-	1(11)	-
BMI (kg/m²)	Mean	24.2	24.5	26.5	27.0	25.8	25.0
	SD	3.24	2.90	3.00	2.50	2.78	5.48
	Range	20.1–30.1	20.3–29.0	21.4–29.7	21.3–29.8	22.2–29.4	20.0–35.5
	Median	25.2	24.0	27.4	27.7	26.8	23.8
Height (cm)	Mean	172	160	178.1	165	166	180
	SD	6.5	4.8	8.9	10.9	9.6	6.1
	Range	164–183	154–170	163–190	150–187	152–185	172–192
	Median	171	158	179	164	166	179
Weight (kg)	Mean	72.0	62.7	83.7[a]	74.3	71.3	81.2
	SD	10.38	10.06	7.31[a]	14.48	9.53	18.81
	Range	56.4–87.5	50.0–80.0	71.6–94.8[a]	48.0–102.0	58–85	59–115
	Median	70.4	60.0	84.8[a]	74.0	71	77

[a]N=4

Pharmacokinetics

Descriptive statistics of PK parameters of TBR-652 are presented in **Table 2**.

Table 2. Mean (CV%) PK and PD Parameters of TBR-652 in Plasma – Day 1 and Day 10

	1 x 25 mg F1 Tablet			1 x 100 mg F2 Tablet		
	Healthy		HIV-1	Healthy		HIV-1
Parameters	652-1-101 (Fasted) N=9	652-1-102 (High Fat) N=9	652-2-201 (Low Fat) N=9	652-1-102 (Low Fat) N=11	652-1-102 (High Fat) N=9	652-2-201 (Low Fat)[d] N=8
Day 1						
C_{max} (ng/mL)	39.4 (23.1)	71.3 (39.0)	30.8 (52.5)	182.3 (65.0)	281.2 (38.0)	179.1 (64.4)
T_{max} (h)[a]	4.00 (3.00–6.00)	3.00 (2.00–6.00)	3.00 (2.00–6.05)	6.00 (3.00–6.00)	6.00 (3.00–6.00)	4.00 (2.08–6.00)
$AUC_{0\text{-}24}$ (ng*h/mL)	407 (23.1)	498 (39.0)	255 (45.1)	1753 (62.9)	2384 (32.0)	1791 (78.5)[c]
C_{max}/Dose	1.58 (23.1)	2.85 (39.0)	1.06 (52.5)	1.75 (69.7)	2.81 (38.0)	1.79 (64.4)
$AUC_{0\text{-}24}$/Dose	16.3 (23.1)	19.9 (39.0)	10.2 (45.1)	17.5 (62.9)	23.8 (32.0)	19.6 (74.7)[c]
CL/F (L/h)	37.5 (22.0)	Missing	Missing	35.9 (17.3–796)[a]	Missing	Missing
$t_{½}$ (h)	35.2 (30.0)	Missing	Missing	35.3 (34.0)	Missing	Missing
Day 10						
C_{min} (ng/mL)	N/A	11.7 (31.0)[b]	6.88 (55.7)	N/A	91.3 (21.0)	32.5 (67.6)[c]
C_{max} (ng/mL)	N/A	65.5 (39.0)[b]	35.1 (46.9)	N/A	381.6 (25.0)	152.6 (35.5)
T_{max} (h)[a]	N/A	3.00 (2.00–4.10)[b]	2.98 (1.00–4.03)	N/A	6.00 (3.00–6.00)	3.92 (3.00–6.00)
$AUC_{0\text{-}24}$ (ng*h/mL)	N/A	605 (43.0)[b]	382 (52.8)	N/A	4142 (22.0)	2088 (37.0)[c]
C_{max}/Dose	N/A	2.62 (39.0)[b]	1.40 (46.9)	N/A	3.81 (25.0)	1.53 (35.5)
$AUC_{0\text{-}24}$/Dose	N/A	24.2 (43.0)[b]	15.3 (52.8)	N/A	41.4 (22.0)	20.9 (37.0)[c]
$t_{½}$ (h)	N/A	40.5 (28.0)[b]	22.5 (42.7)	N/A	46.2 (22.0)	33.0 (24.3)
Accumulation Ratio[a]	N/A	1.39 (0.753–2.09)[b]	1.56 (0.729–3.37)	N/A	1.62 (1.33–4.29)	1.60 (0.351–236)
CL/F (L/h)	N/A	47.7 (38.2)[b]	81.3 (45.3)	N/A	25.2 (22.7)	55.0 (40.9)

N/A: Not available. Multiple once-daily administrations of TBR-652 were not investigated.
Missing: Due to the once-daily administration, the terminal elimination phase was not discernible on Day 1.
[a]Median (Min–Max); [b]N=8; [c]N=7; [d]Descriptive statistics for the 100 mg dose group were computed without two patients who were dosed in Argentina. These Argentinean patients displayed modest differences in mean PK profiles and parameters that may have been related to an altered exposure to TBR-652 resulting from the co-administration with a meal of higher-fat content relative to the patients dosed in the US clinics.

Summary concentration profiles of TBR-652 in plasma following single (**Figures 1 and 2**) and once daily dosing for 10 days (**Figures 3 and 4**) are depicted.

Figure 1. Mean TBR-652 Concentration-Time Profiles in Plasma Following a Single Administration of 1 x 25 mg F1 Tablets—Day 1

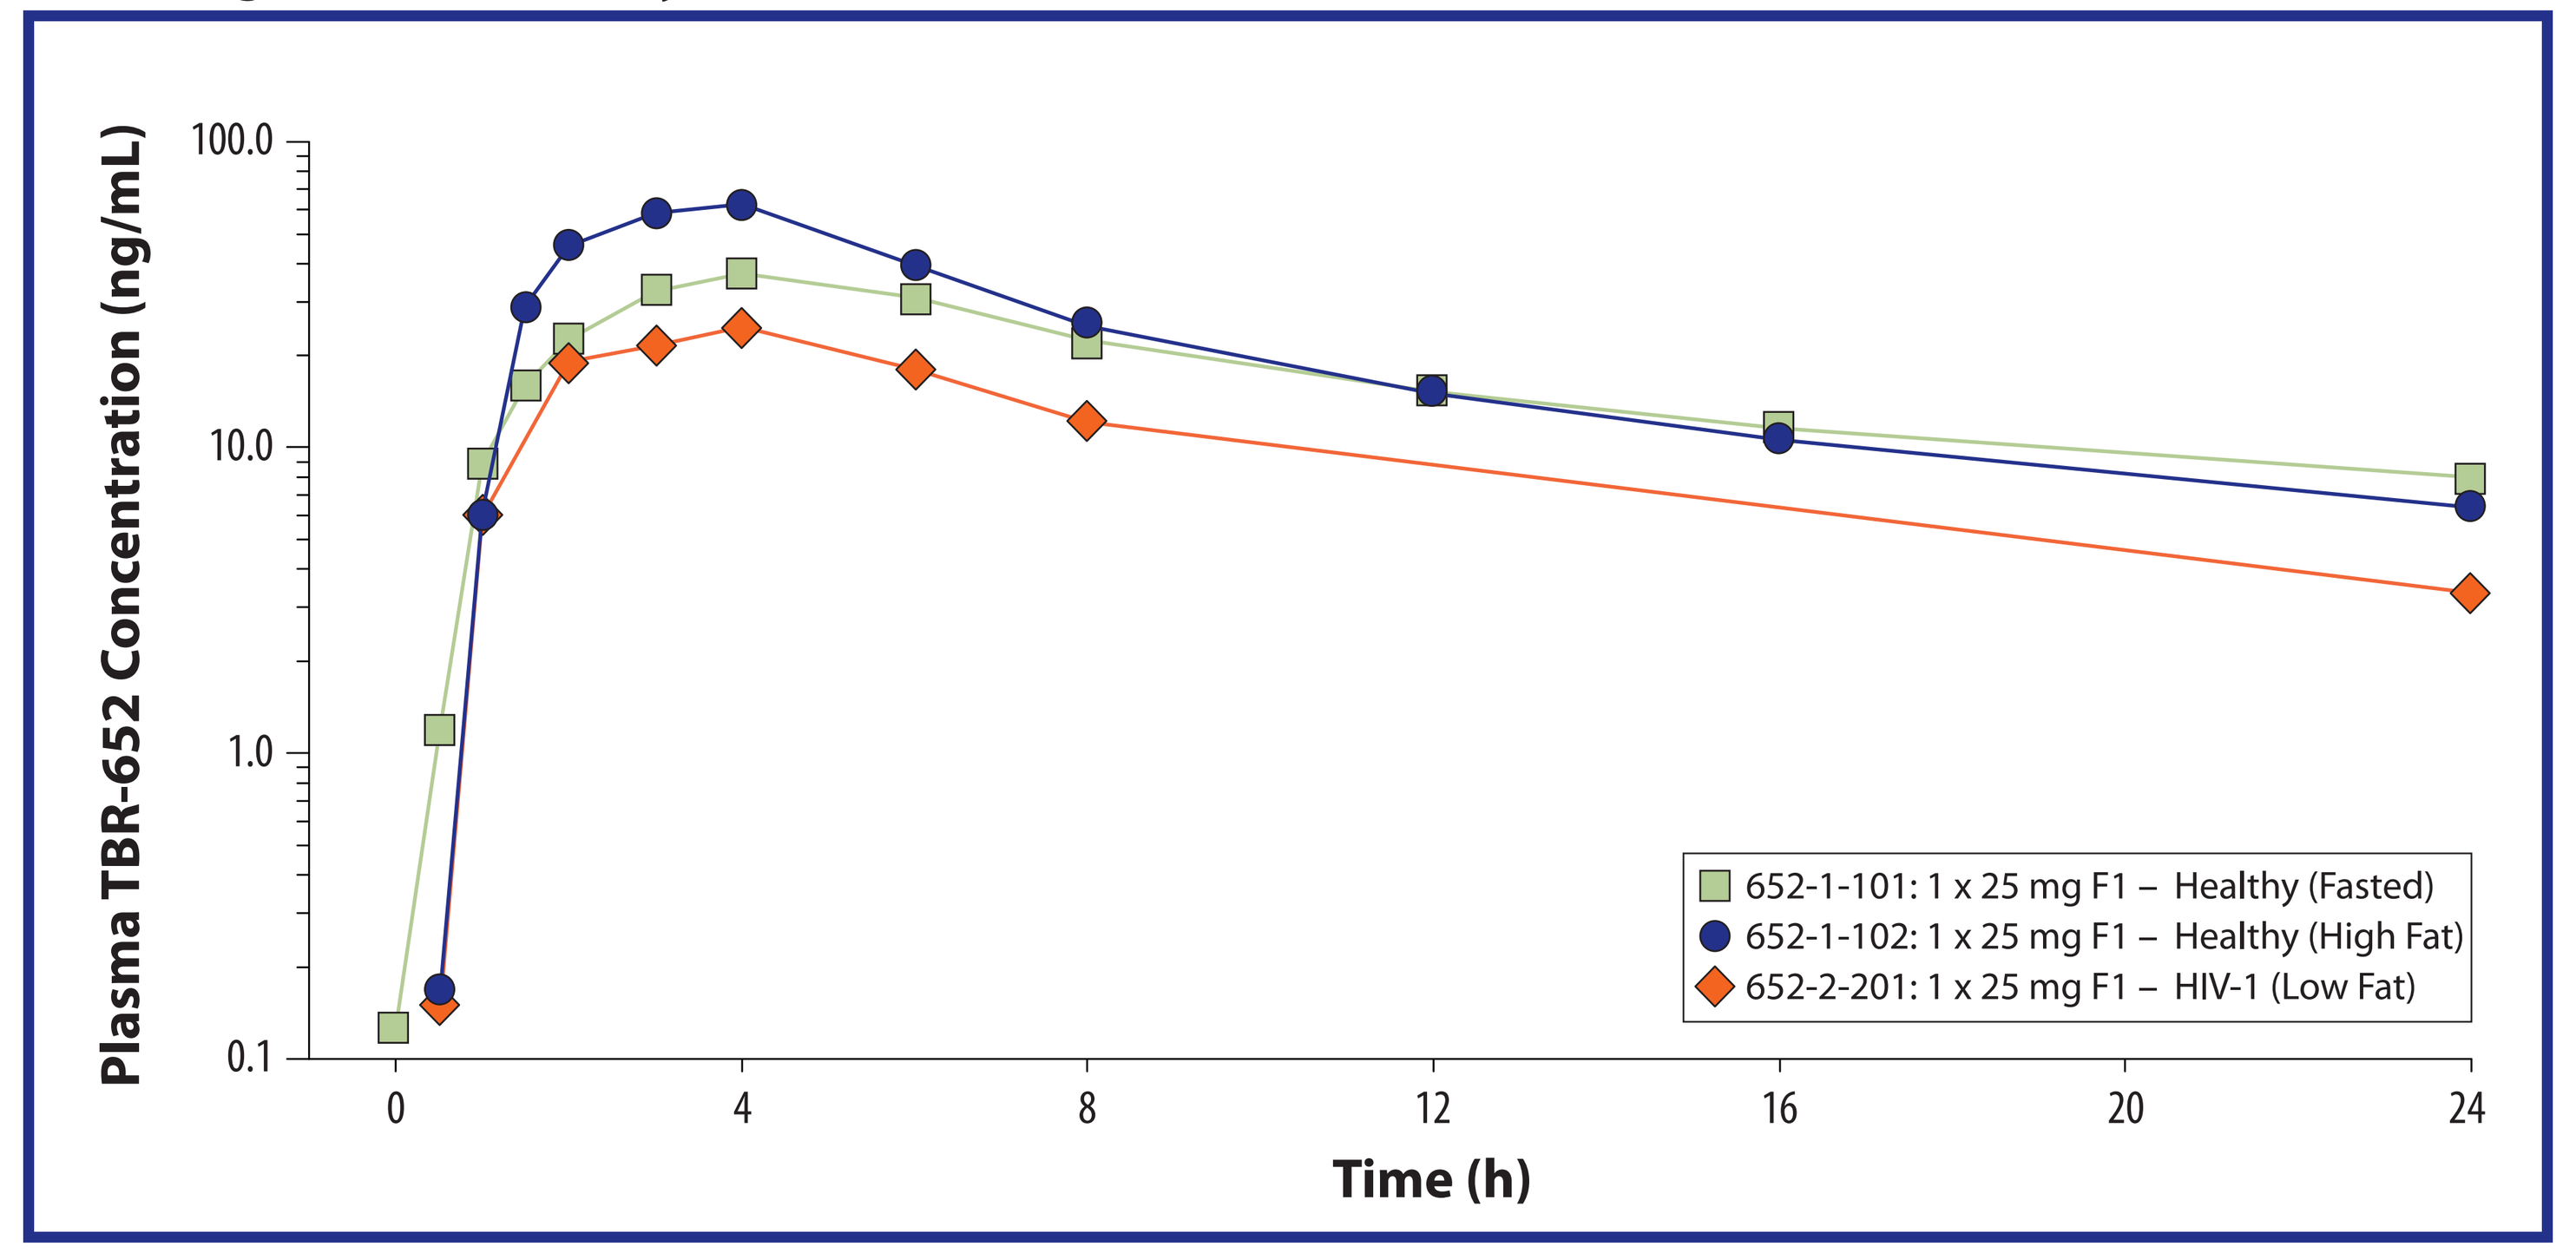
Plasma TBR-652 Concentration (ng/mL)
Time (h)
652-1-101: 1 x 25 mg F1 – Healthy (Fasted)
652-1-102: 1 x 25 mg F1 – Healthy (High Fat)
652-2-201: 1 x 25 mg F1 – HIV-1 (Low Fat)

Figure 2. Mean TBR-652 Concentration-Time Profiles in Plasma Following a Single Administration of 1 x 100 mg F2 Tablets—Day 1

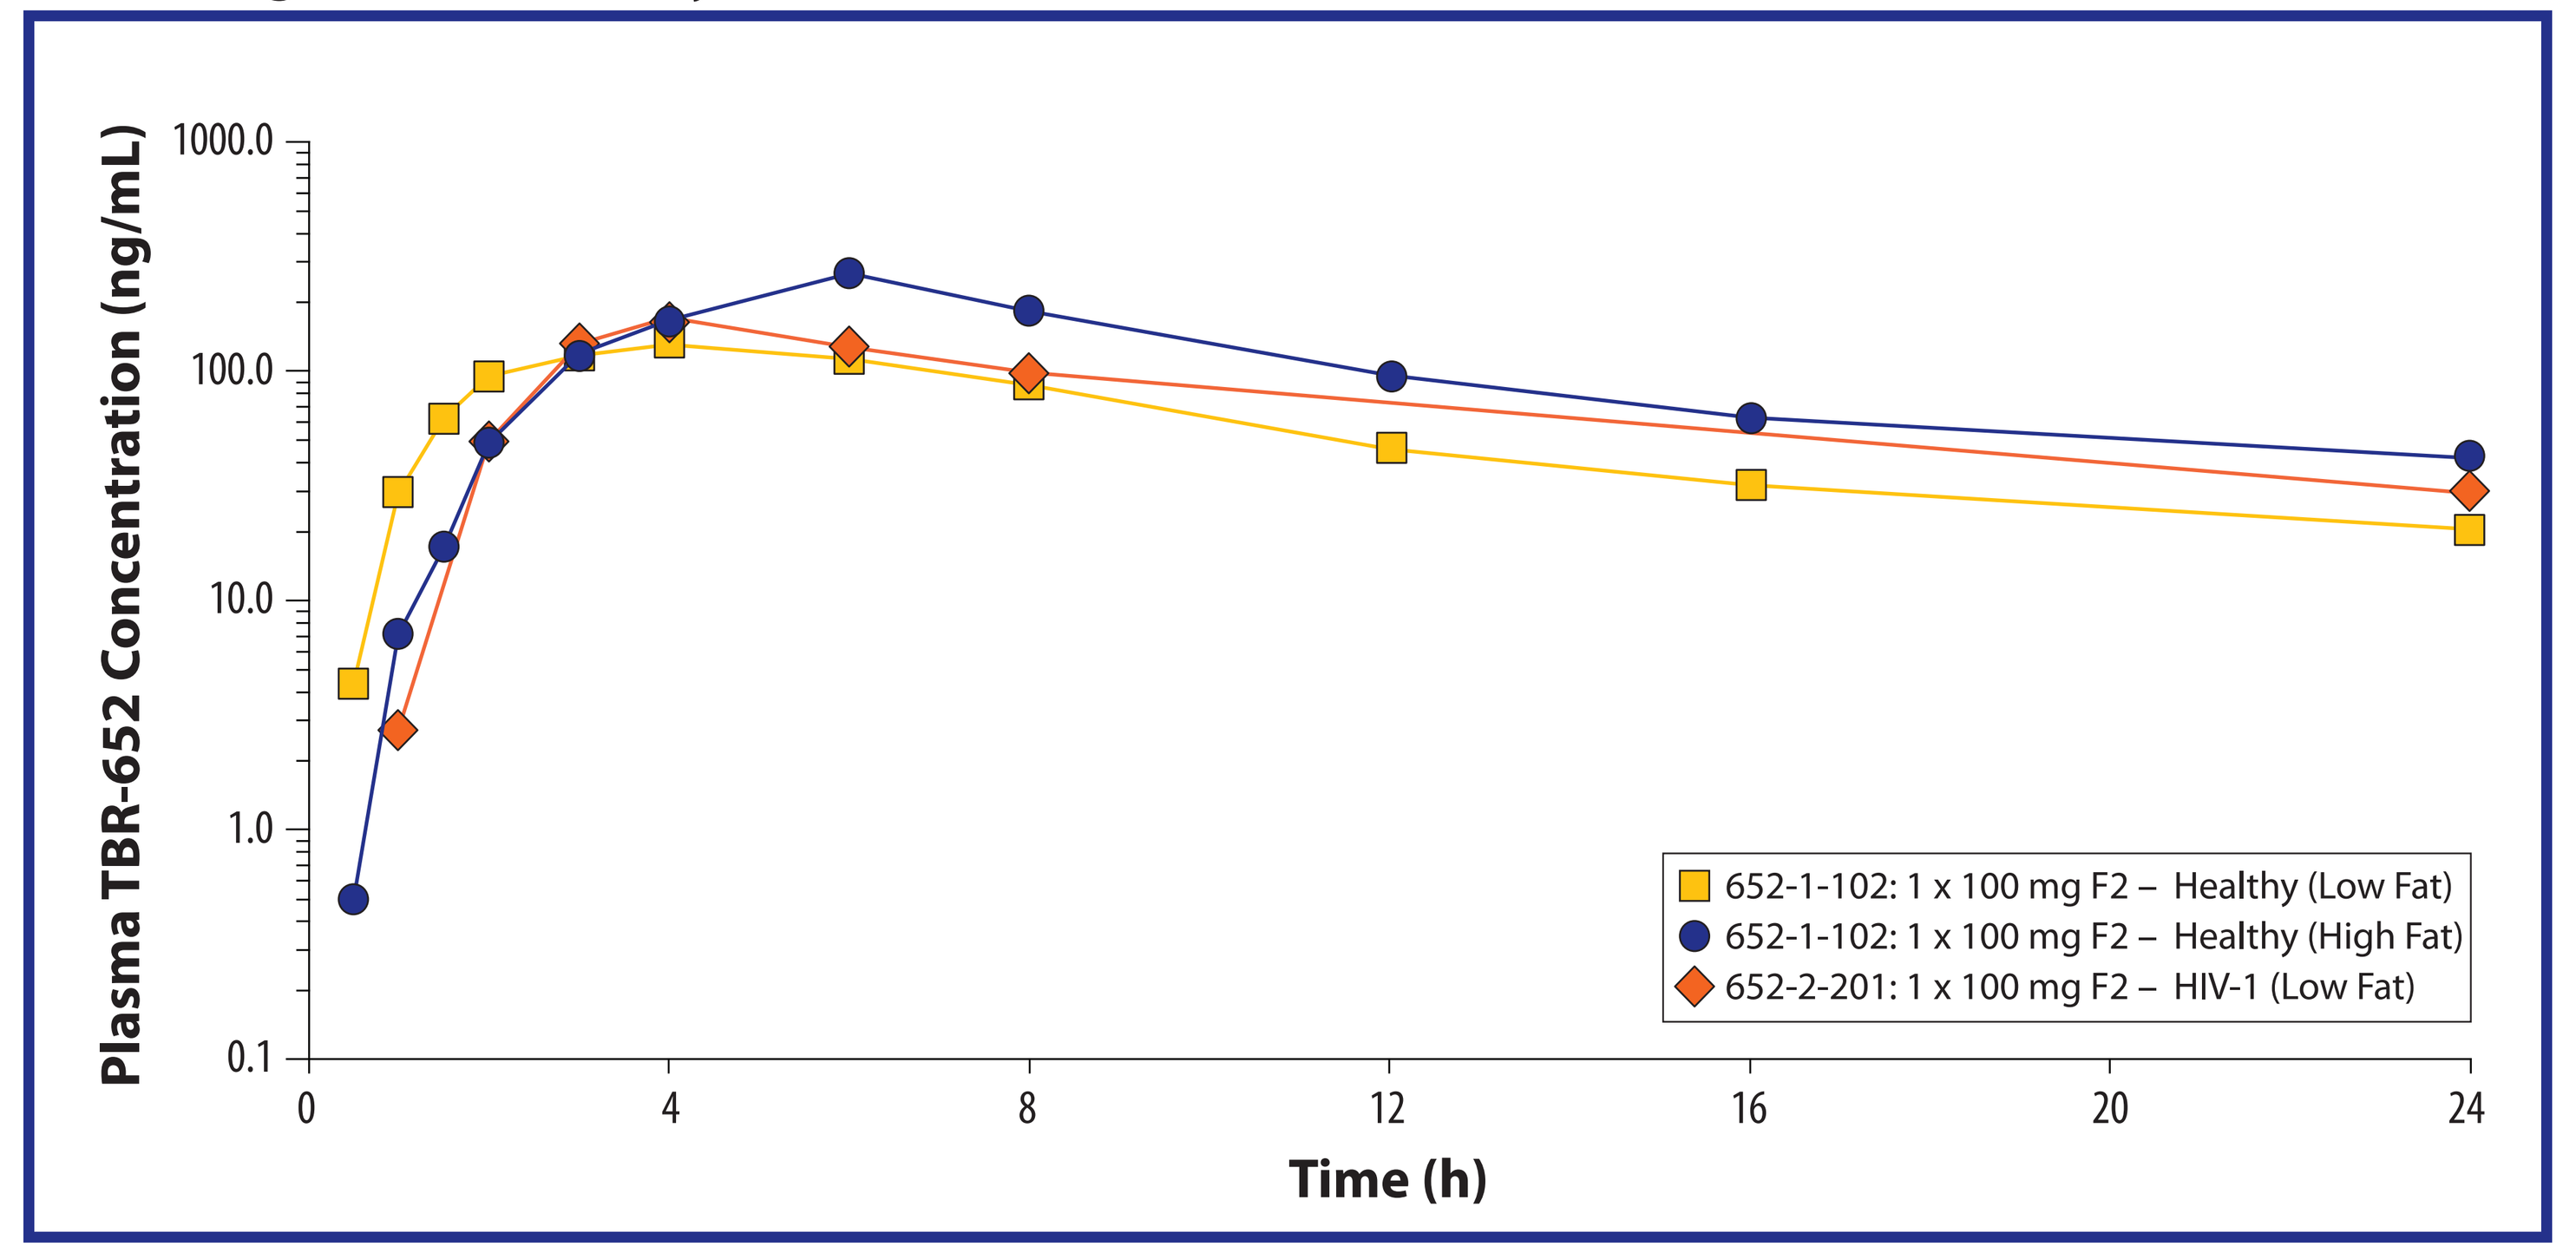
Plasma TBR-652 Concentration (ng/mL)
Time (h)
652-1-102: 1 x 100 mg F2 – Healthy (Low Fat)
652-1-102: 1 x 100 mg F2 – Healthy (High Fat)
652-2-201: 1 x 100 mg F2 – HIV-1 (Low Fat)

Figure 3. Mean TBR-652 Concentration-Time Profiles in Plasma Following Oral QD TBR-652 (1 x 25 mg F1 Tablets) Administrations for 10 Days—Semi-Log Plot

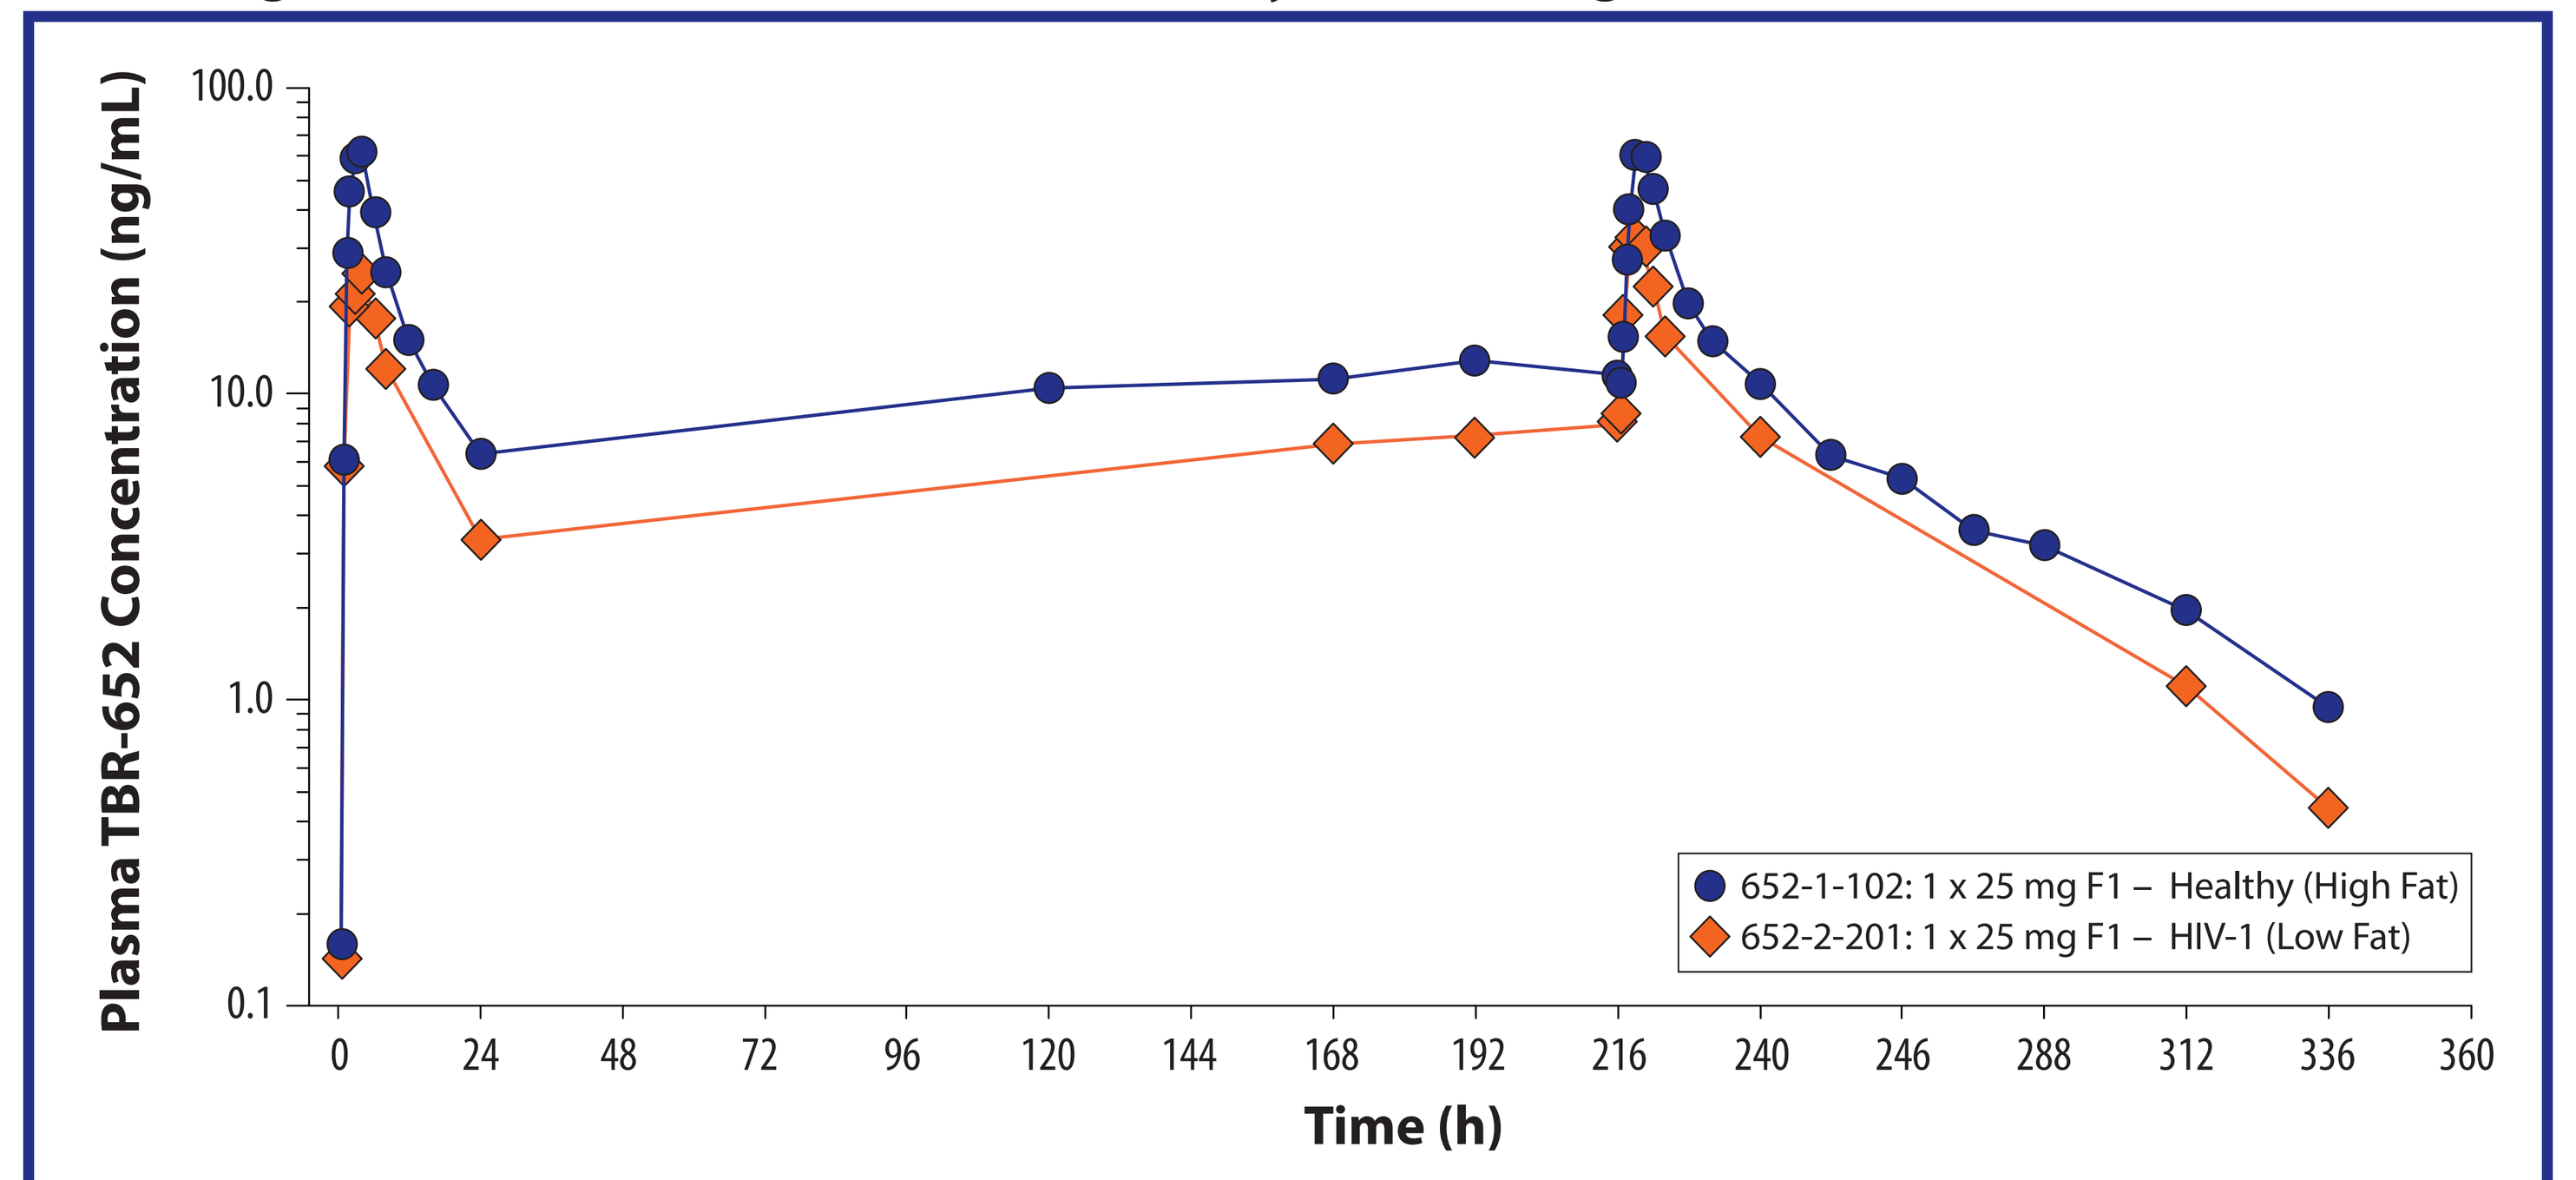
Plasma TBR-652 Concentration (ng/mL)
Time (h)
652-1-102: 1 x 25 mg F1 – Healthy (High Fat)
652-2-201: 1 x 25 mg F1 – HIV-1 (Low Fat)

Multiple once-daily administrations of 25 mg F1 tablets of TBR-652 were not investigated in healthy subjects under fasted conditions.

Figure 4. Mean TBR-652 Concentration-Time Profiles in Plasma Following Oral QD TBR-652 (1 x 100 mg F2 Tablets) Administrations for 10 Days—Semi-Log Plot

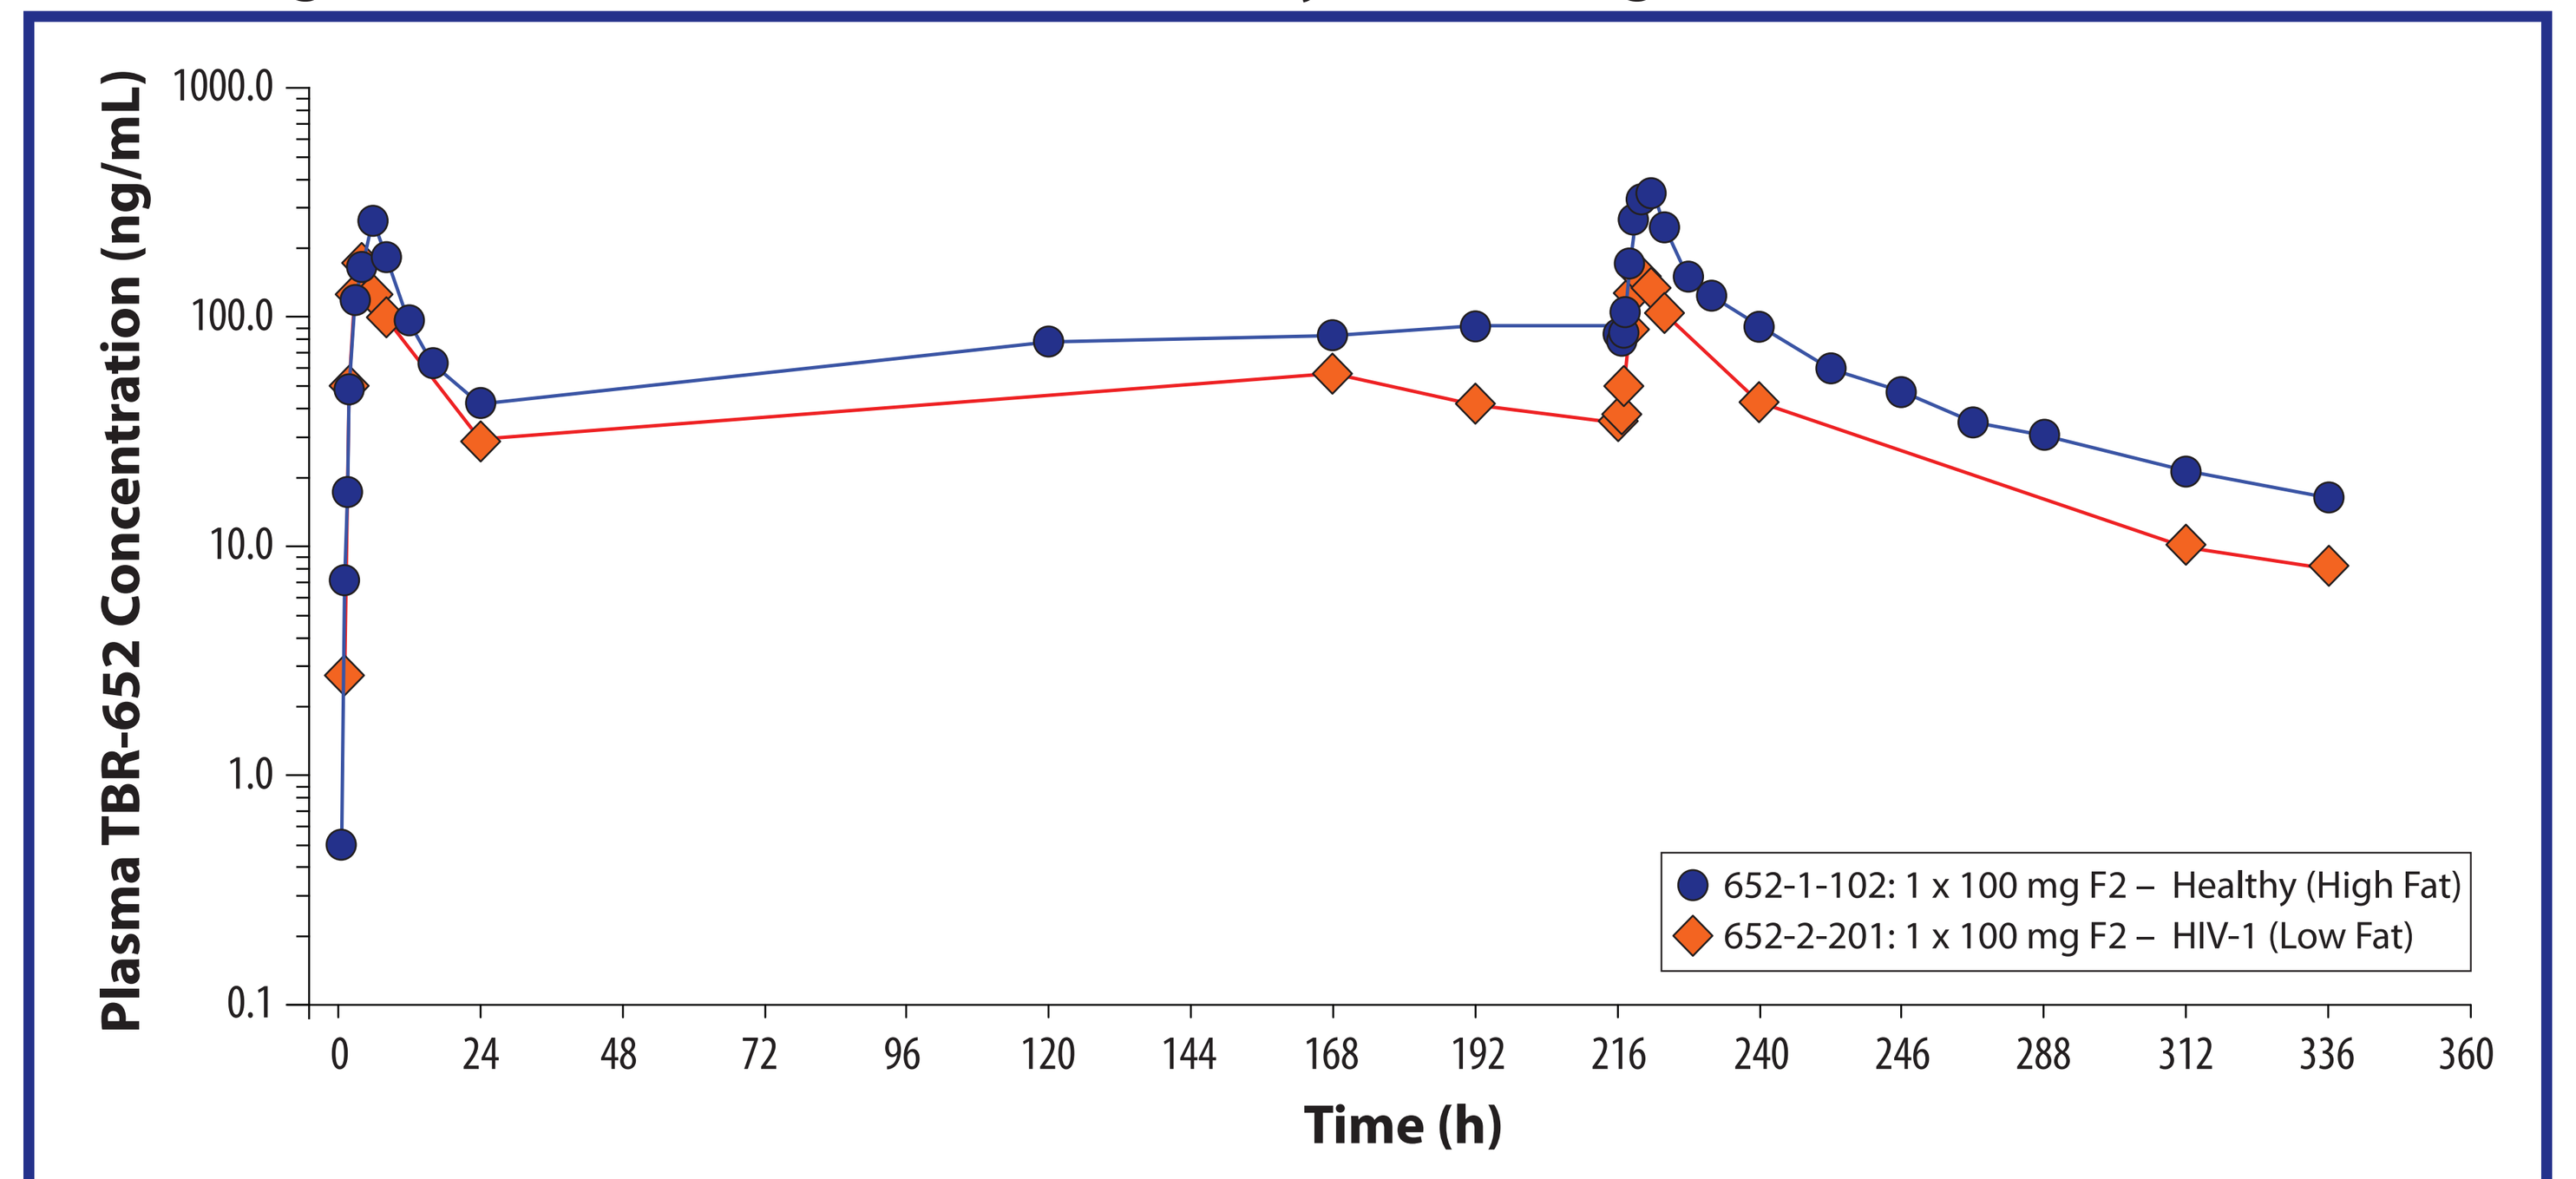
Plasma TBR-652 Concentration (ng/mL)
Time (h)
652-1-102: 1 x 100 mg F2 – Healthy (High Fat)
652-2-201: 1 x 100 mg F2 – HIV-1 (Low Fat)

Multiple once-daily administrations of 100 mg F2 tablets of TBR-652 were not investigated in healthy subjects under fasted conditions, or with a high-fat meal.

Figure 5. Mean Dose-Normalized TBR-652 $AUC_{0\text{-}24}$ in Plasma Following a Single Oral TBR-652 (25 mg F1 Tablets)—Semi-Log Plot

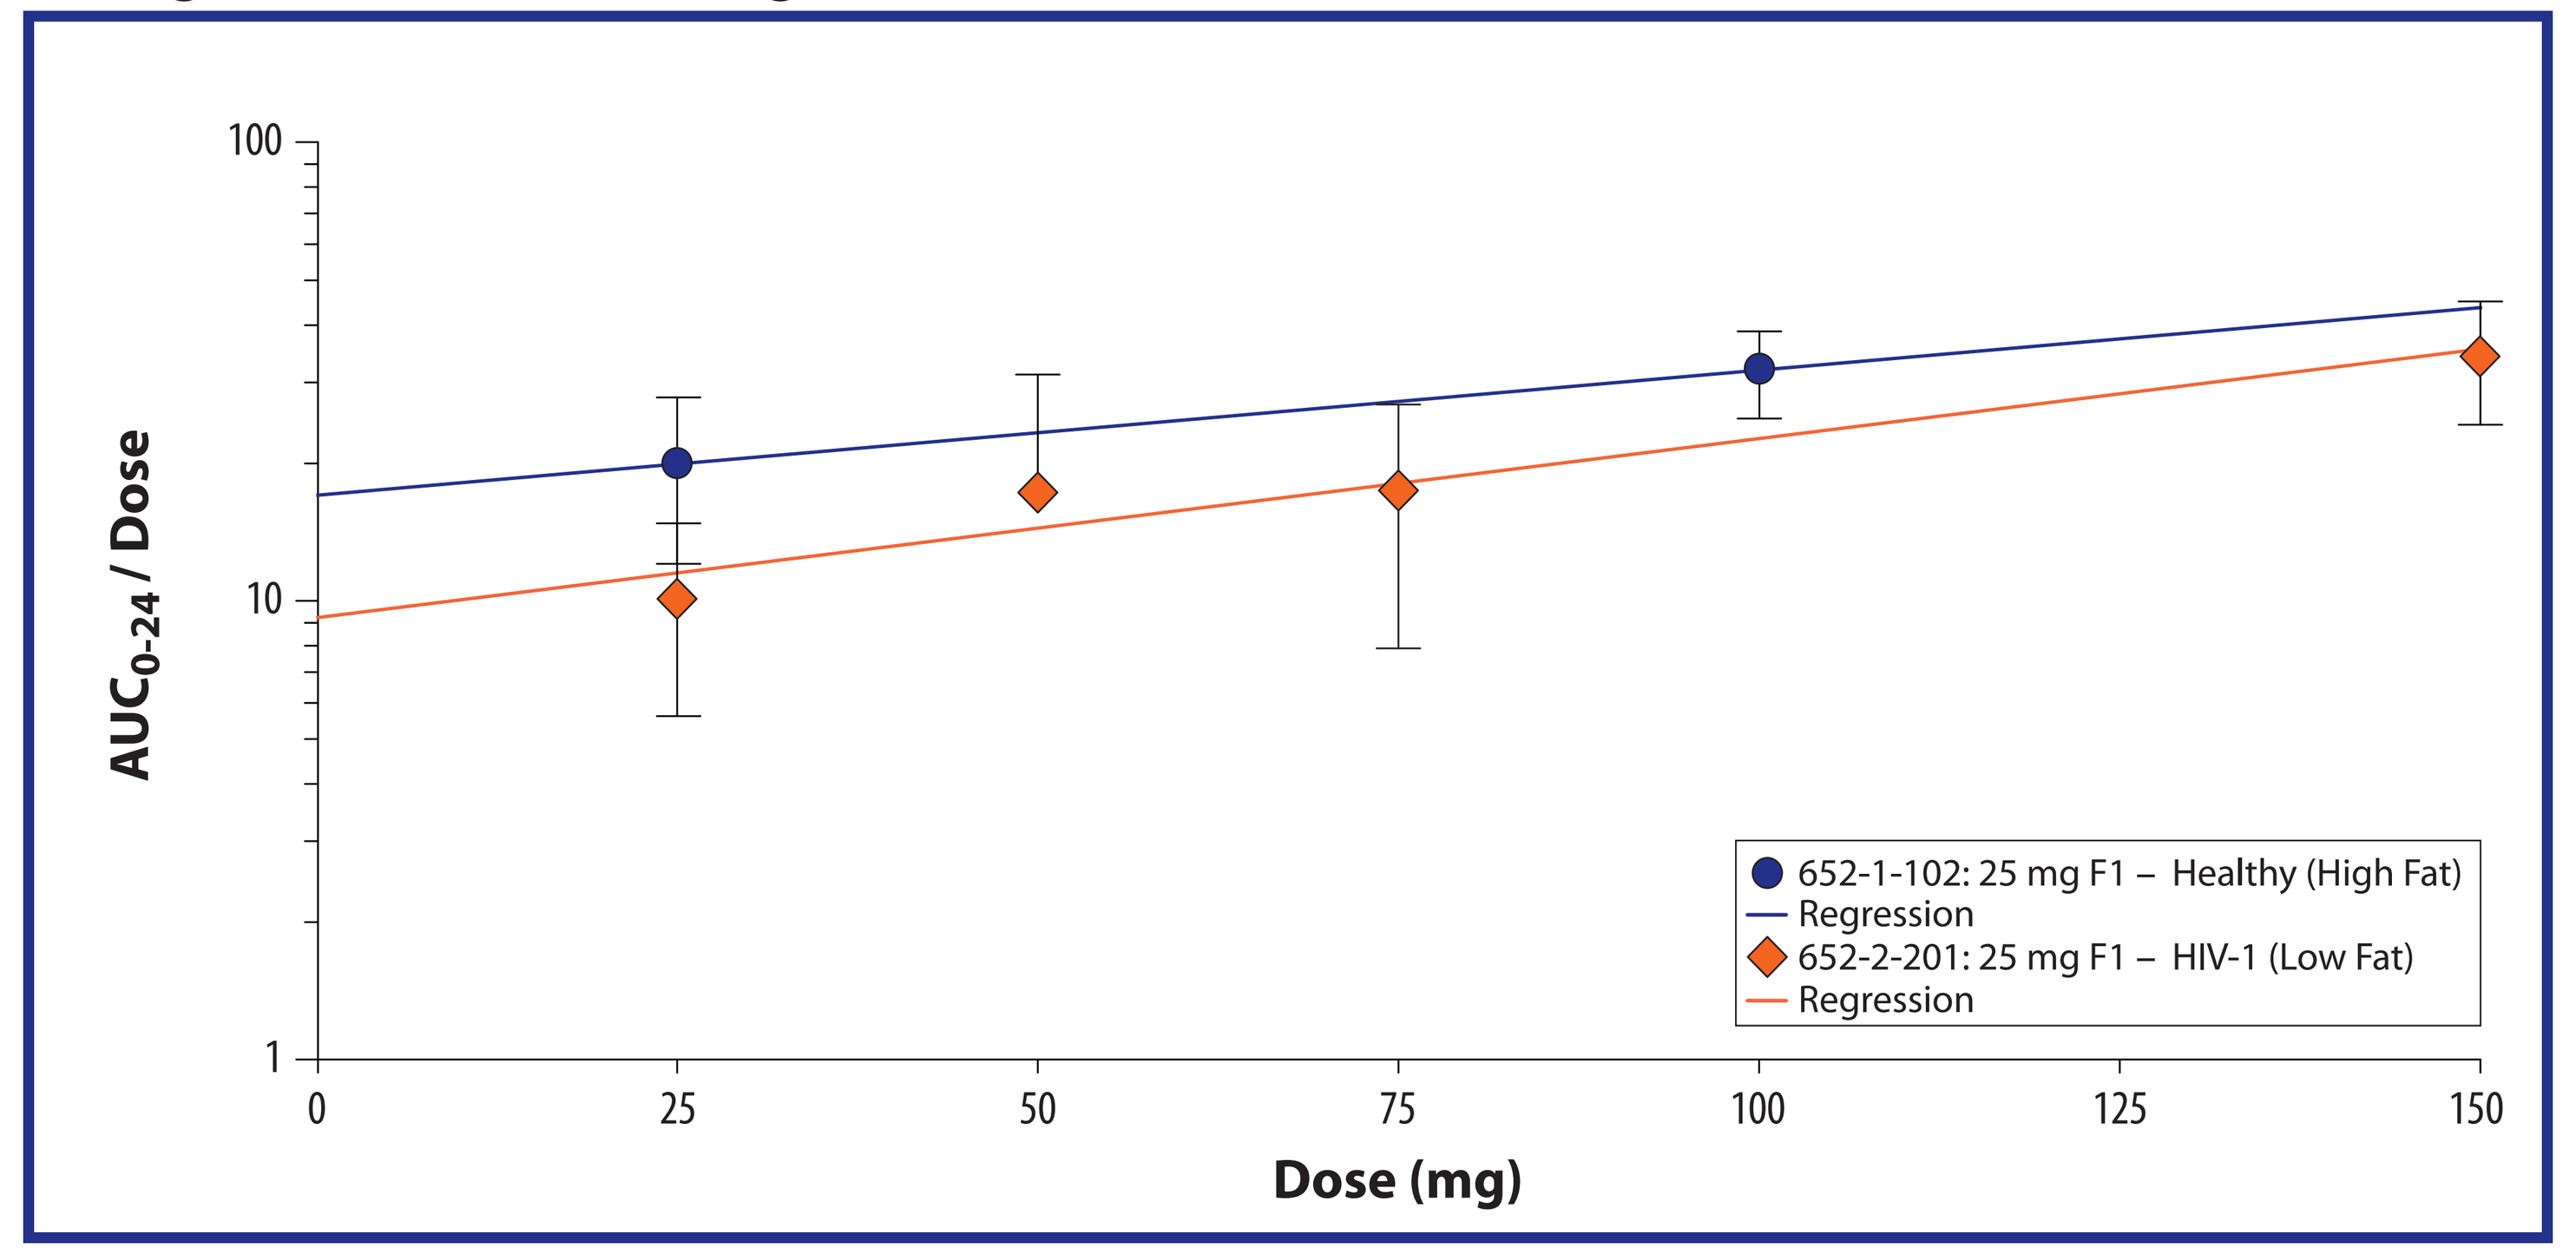
AUC0-24 / Dose
Dose (mg)
652-1-102: 25 mg F1 – Healthy (High Fat)
Regression
652-2-201: 25 mg F1 – HIV-1 (Low Fat)
Regression

Figure 6. Mean Dose-Normalized TBR-652 C_{max} in Plasma Following a Single Oral TBR-652 (25 mg F1 Tablets)—Semi-Log Plot

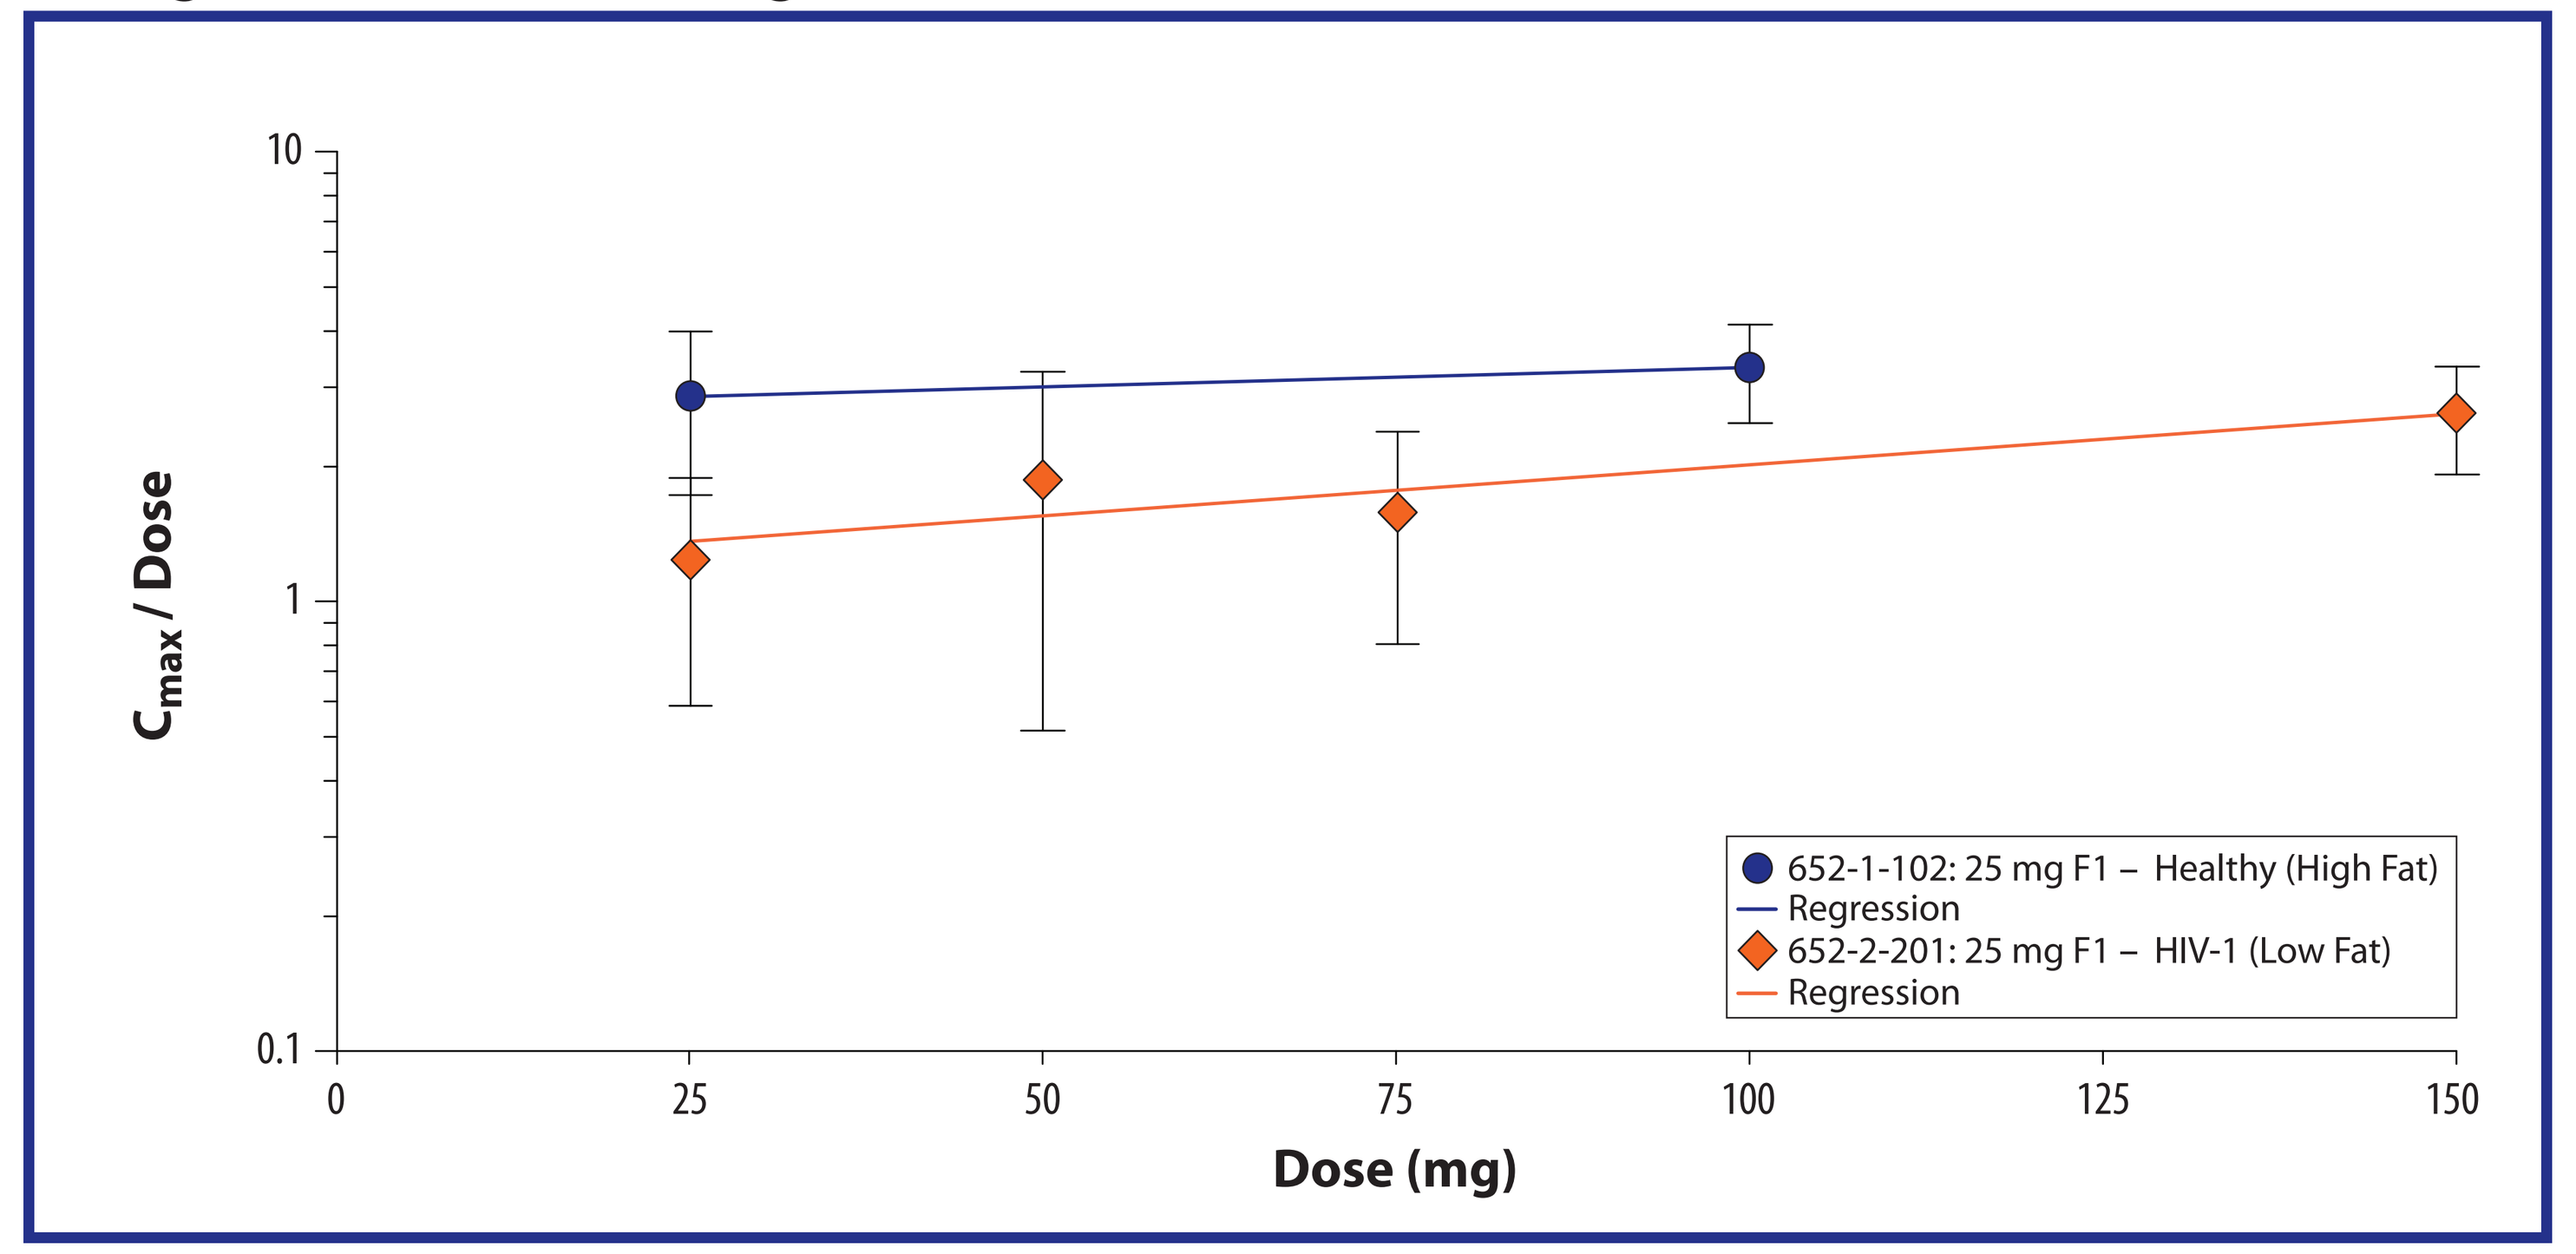
Cmax / Dose
Dose (mg)
652-1-102: 25 mg F1 – Healthy (High Fat)
Regression
652-2-201: 25 mg F1 – HIV-1 (Low Fat)
Regression

DISCUSSION

- In healthy and HIV-1 infected subjects, TBR-652 was well absorbed following single and repeated dosing of TBR-652 25 and 100 mg QD.
- Slight variations were observed in the exposure of TBR-652 between healthy and HIV-1 infected subjects which was attributed to differences in meal content. Results from the food effect study[3] showed that a high-fat meal significantly increases the extent of TBR-652 bioavailability and reduces the intersubject variability in plasma TBR-652 concentrations.[4]
- Single dose administration of the 25 mg F1 tablets of TBR-652 under fed conditions resulted in a dose-proportional increase in $AUC_{0\text{-}24}$ and C_{max} over the dose range studied (10 to 100 mg QD) in healthy and HIV-1 infected subjects. The dose proportionality of TBR-652 in plasma for healthy subjects under fasted conditions has been previously presented.[2]
- Median accumulation ratios for TBR-652 $AUC_{0\text{-}24}$ in healthy and HIV-1 infected subjects following repeated once-daily doses for the 25 and 100 mg QD doses were 1.4 to 1.6, respectively. These results suggest a moderate accumulation of TBR-652 in healthy and HIV-1 infected patients following repeated dosing.
- The elimination kinetics of TBR-652 were similar in healthy and HIV-1 infected subjects across the dose range studied, with mean $t_{½}$ values ranging from 23 to 46 hours.

CONCLUSIONS

- TBR-652 was well absorbed following repeated dosing of 25 and 100 mg QD.
- The PK of TBR-652 in healthy subjects was similar to that observed in HIV-1 infected subjects under fed and fasted conditions, and systemic exposure to TBR-652 increased in a dose proportional manner from 25 to100 mg QD.
- The data from these studies support the further development of TBR-652 in HIV-1 infected patients. Plans for new formulation and Phase 2b clinical development are underway.

REFERENCES

1. Lalezari J, Gathe J, Thompson M, Cohen C, DeJesus E, Brinson C, Elion R, Galindez J, Palleja S, Martin D. Safety and Efficacy of TBR-652, a Chemokine Receptor 5 (CCR5) Antagonist, in HIV-1-Infected, Antiretroviral (ARV) Treatment-Experienced, CCR5 Antagonist–Naïve Patients. Presented at the 17th Conference on Retroviruses and Opportunistic Infections. San Francisco, CA, February 16-19, 2010. Oral presentation K-111.
2. Palleja S, Driz R, Wang-Smith L, Ogden R, Martin D, Bobbitt A, Sapirstein J. Pharmacokinetics (PK), safety, and tolerability of single doses of the chemokine C-C receptor 5 (CCR5) antagonist TBR-652 in healthy volunteers. Presented at the 5th IAS Conference on HIV Pathogenesis and Treatment. Cape Town, South Africa, July 19-22, 2009. Poster #WEPEB251.
3. Palleja S, Wang-Smith L, Ogden R, Martin D, Driz R, Sapirstein J. TBR-652, a Chemokine Receptor 5 (CCR5) Antagonist, Demonstrates Good Oral Bioavailability and Desirable Pharmacokinetic (PK) and Safety Profiles in Healthy Volunteers. Presented at the 49th Annual ICAAC Annual Meeting, San Francisco, CA, September 12-15, 2009. Poster #A1-1308.
4. Martin D, Palleja S, Pheng L, Trinh M, Marier JF, Sapirstein J. Pharmacokinetics (PK) and Pharmacodynamics (PD) of TBR-652, a Chemokine Receptor 5 (CCR5) Antagonist, in HIV 1-Infected, Antiretroviral (ARV) Treatment-Experienced, CCR5 Antagonist–Naïve Patients. Presented at the 17th Conference on Retroviruses and Opportunistic Infections. San Francisco, CA, February 16-19, 2010. Poster 598.

ACKNOWLEDGEMENTS

Thank you to the investigators, the patients, and all the study personnel.

The 50th Interscience Conference on Antimicrobial Agents and Chemotherapy
September 12-15, 2010 in Boston, Massachusetts