598

PHARMACOKINETICS (PK) AND PHARMACODYNAMICS (PD) OF TBR-652, A CHEMOKINE RECEPTOR 5 (CCR5) ANTAGONIST, IN HIV-1–INFECTED, ANTIRETROVIRAL (ARV) TREATMENT-EXPERIENCED, CCR5 ANTAGONIST-NAÏVE PATIENTS

David Martin[1], S Palleja[1], L Pheng[2], M Trinh[2], JF Marier[2], J Sapirstein[1] • [1]Tobira Therapeutics, Inc., Princeton, NJ, USA; [2]Pharsight, a Certara Company, Montreal, Quebec, Canada

David Martin, PharmD
214 Carnegie Center, Suite 306
Princeton, NJ 08540
(609) 897-1102
dmartin@tobiratherapeutics.com

INTRODUCTION

Early studies with compounds designed to block CCR5 have shown CCR5 antagonists to be effective in preventing HIV-1 cell entry and subsequent replication when administered in combination with other antiretroviral drugs. TBR-652, a promising CCR5 antagonist, was well tolerated in initial healthy volunteer studies, where phase 1 pharmacokinetic (PK) studies showed the feasibility of once-daily oral dosing. This is the first report of TBR-652 PK and pharmacodynamics (PD) in HIV-1–infected patients.

OBJECTIVES

To determine the PK, dose response, and antiviral activity of TBR-652 monotherapy at 5 dose levels when administered orally once daily (QD) for 10 days in HIV-1–infected, antiretroviral treatment-experienced patients.

METHODS

Study Design

This was a double-blind, placebo-controlled, randomized (4:1 TBR-652: placebo), dose-escalating study to assess the PK/PD of TBR-652 monotherapy given orally QD for 10 days in a total of 54 patients. Patients had plasma HIV-1 RNA levels ≥5000 copies/mL, CD4+ cell counts ≥250 cells/mm^3, and no CXCR4 or dual/mixed tropism. All patients in a cohort completed 10 days of dosing prior to dose escalation. PK collections were on Days 1 and 10 at predose (time 0), and at 0.5, 1, 2, 3, 4, 6, 8, and 24 hours postdose; on Days 2–9 at predose (trough samples); and on Days 14 and 15 (96 and 120 hours post last dose). HIV-1 RNA collections were at Day 1 predose and Days 2, 3, 4, 7, 8, 10, 11, 15, 24, and 40.

Analyses

In the 100-mg dose group, analyses were done with and without the data from the only 2 Argentinean patients. They displayed >2-fold increased exposure to TBR-652, attributed to a much higher fat breakfast than everyone else.

Steady-State Analysis

Differences in trough plasma concentrations of TBR-652 on Days 8, 9, and 10 were analyzed using Helmert contrasts deduced from a repeated measurement analysis of variance (ANOVA) model in order to determine the time to steady state.

PK

A noncompartmental PK analysis was performed on the concentration-time profiles of TBR-652 to calculate $AUC_{0\text{-}24}$, $AUC_{0\text{-last}}$, T_{max}, C_{max} λ_z, $t_{½}$, CL/F, and/or V_{ss}/F for all treatments following single and multiple QD dosing of TBR-652. Actual sampling times were used in the calculations of PK parameters. A Power Model[1] was used to evaluate the dose-proportionality of TBR-652. The statistical linear relationship between In-transformed PK parameters $AUC_{0\text{-}24}$ and C_{max} and the In-transformed dose on Days 1 and 10 was assessed.

PD

Antiviral activity and viral pharmacodynamic assessments were performed using actual time points (days) calculated relative to dose or placebo. The following PD parameters were calculated for each plasma HIV-1 RNA vs time profile: change in plasma log HIV-1 RNA from baseline over the 10-day treatment period (ΔLog HIV-1 RNA) and during the follow-up period and plasma HIV-1 RNA change from baseline to nadir (maximum change) (ΔLog HIV-1 RNA_{max}). The baseline HIV-1 RNA level corresponded to the observation collected prior to dosing on Study Day 1.

PK/PD

To characterize the relationship between antiviral activity and drug exposure, E_{max} and linear models were tested.

The relationship between the $AUC_{0\text{-}24}$, C_{max}, and C_{ss} values on Day 10 with the HIV-1 RNA pre-dose observations on Day 11 were described using linear and inhibitory E_{max} models:

Linear Model: $(E = \alpha + \beta * In(C))$ **E_{max} Model:** $E = E_0 + \dfrac{(I_{max} - E_0) \cdot C^{\gamma}}{IC_{50}^{\gamma} + C^{\gamma}}$

Where E is effect, E_0 is the baseline effect (fixed to 0), I_{max} is the maximal inhibition, C denotes the PK variable ($AUC_{0\text{-}24}$, C_{max}, or C_{ss}), IC_{50} is the value of the PK variable which corresponds to 50% of the maximal inhibition and γ is the shape parameter which describes the degree of sigmoidicity. Model selection was based on the minimization of the Akaike Information Criteria (AIC).

RESULTS

Disposition

Fifty-four patients were accrued. Plasma concentration-time profiles of TBR-652 from a total of 43 patients were available for the PK assessment. Plasma HIV-1 RNA from a total of 42 (TBR-652) and 10 (placebo) patients were available for the PD assessment. One patient randomized to the 50 mg/day group received only 25 mg/day and was therefore analyzed in the 25-mg group. One patient received only a single dose, and one patient had dual/mixed tropism at baseline and both did not qualify for the PK and/or PD analyses.

Pharmacokinetics

A summary of noncompartmental analysis PK and PD parameters are presented in **Table 1**. Summary concentration profiles of TBR-652 in plasma following once-daily dosing for 10 days are presented in **Figure 1**. Median plasma viral load changes by treatment are presented in **Figure 2**.

Figure 1. Mean TBR-652 Concentration-Time Profiles in Plasma—Day 10

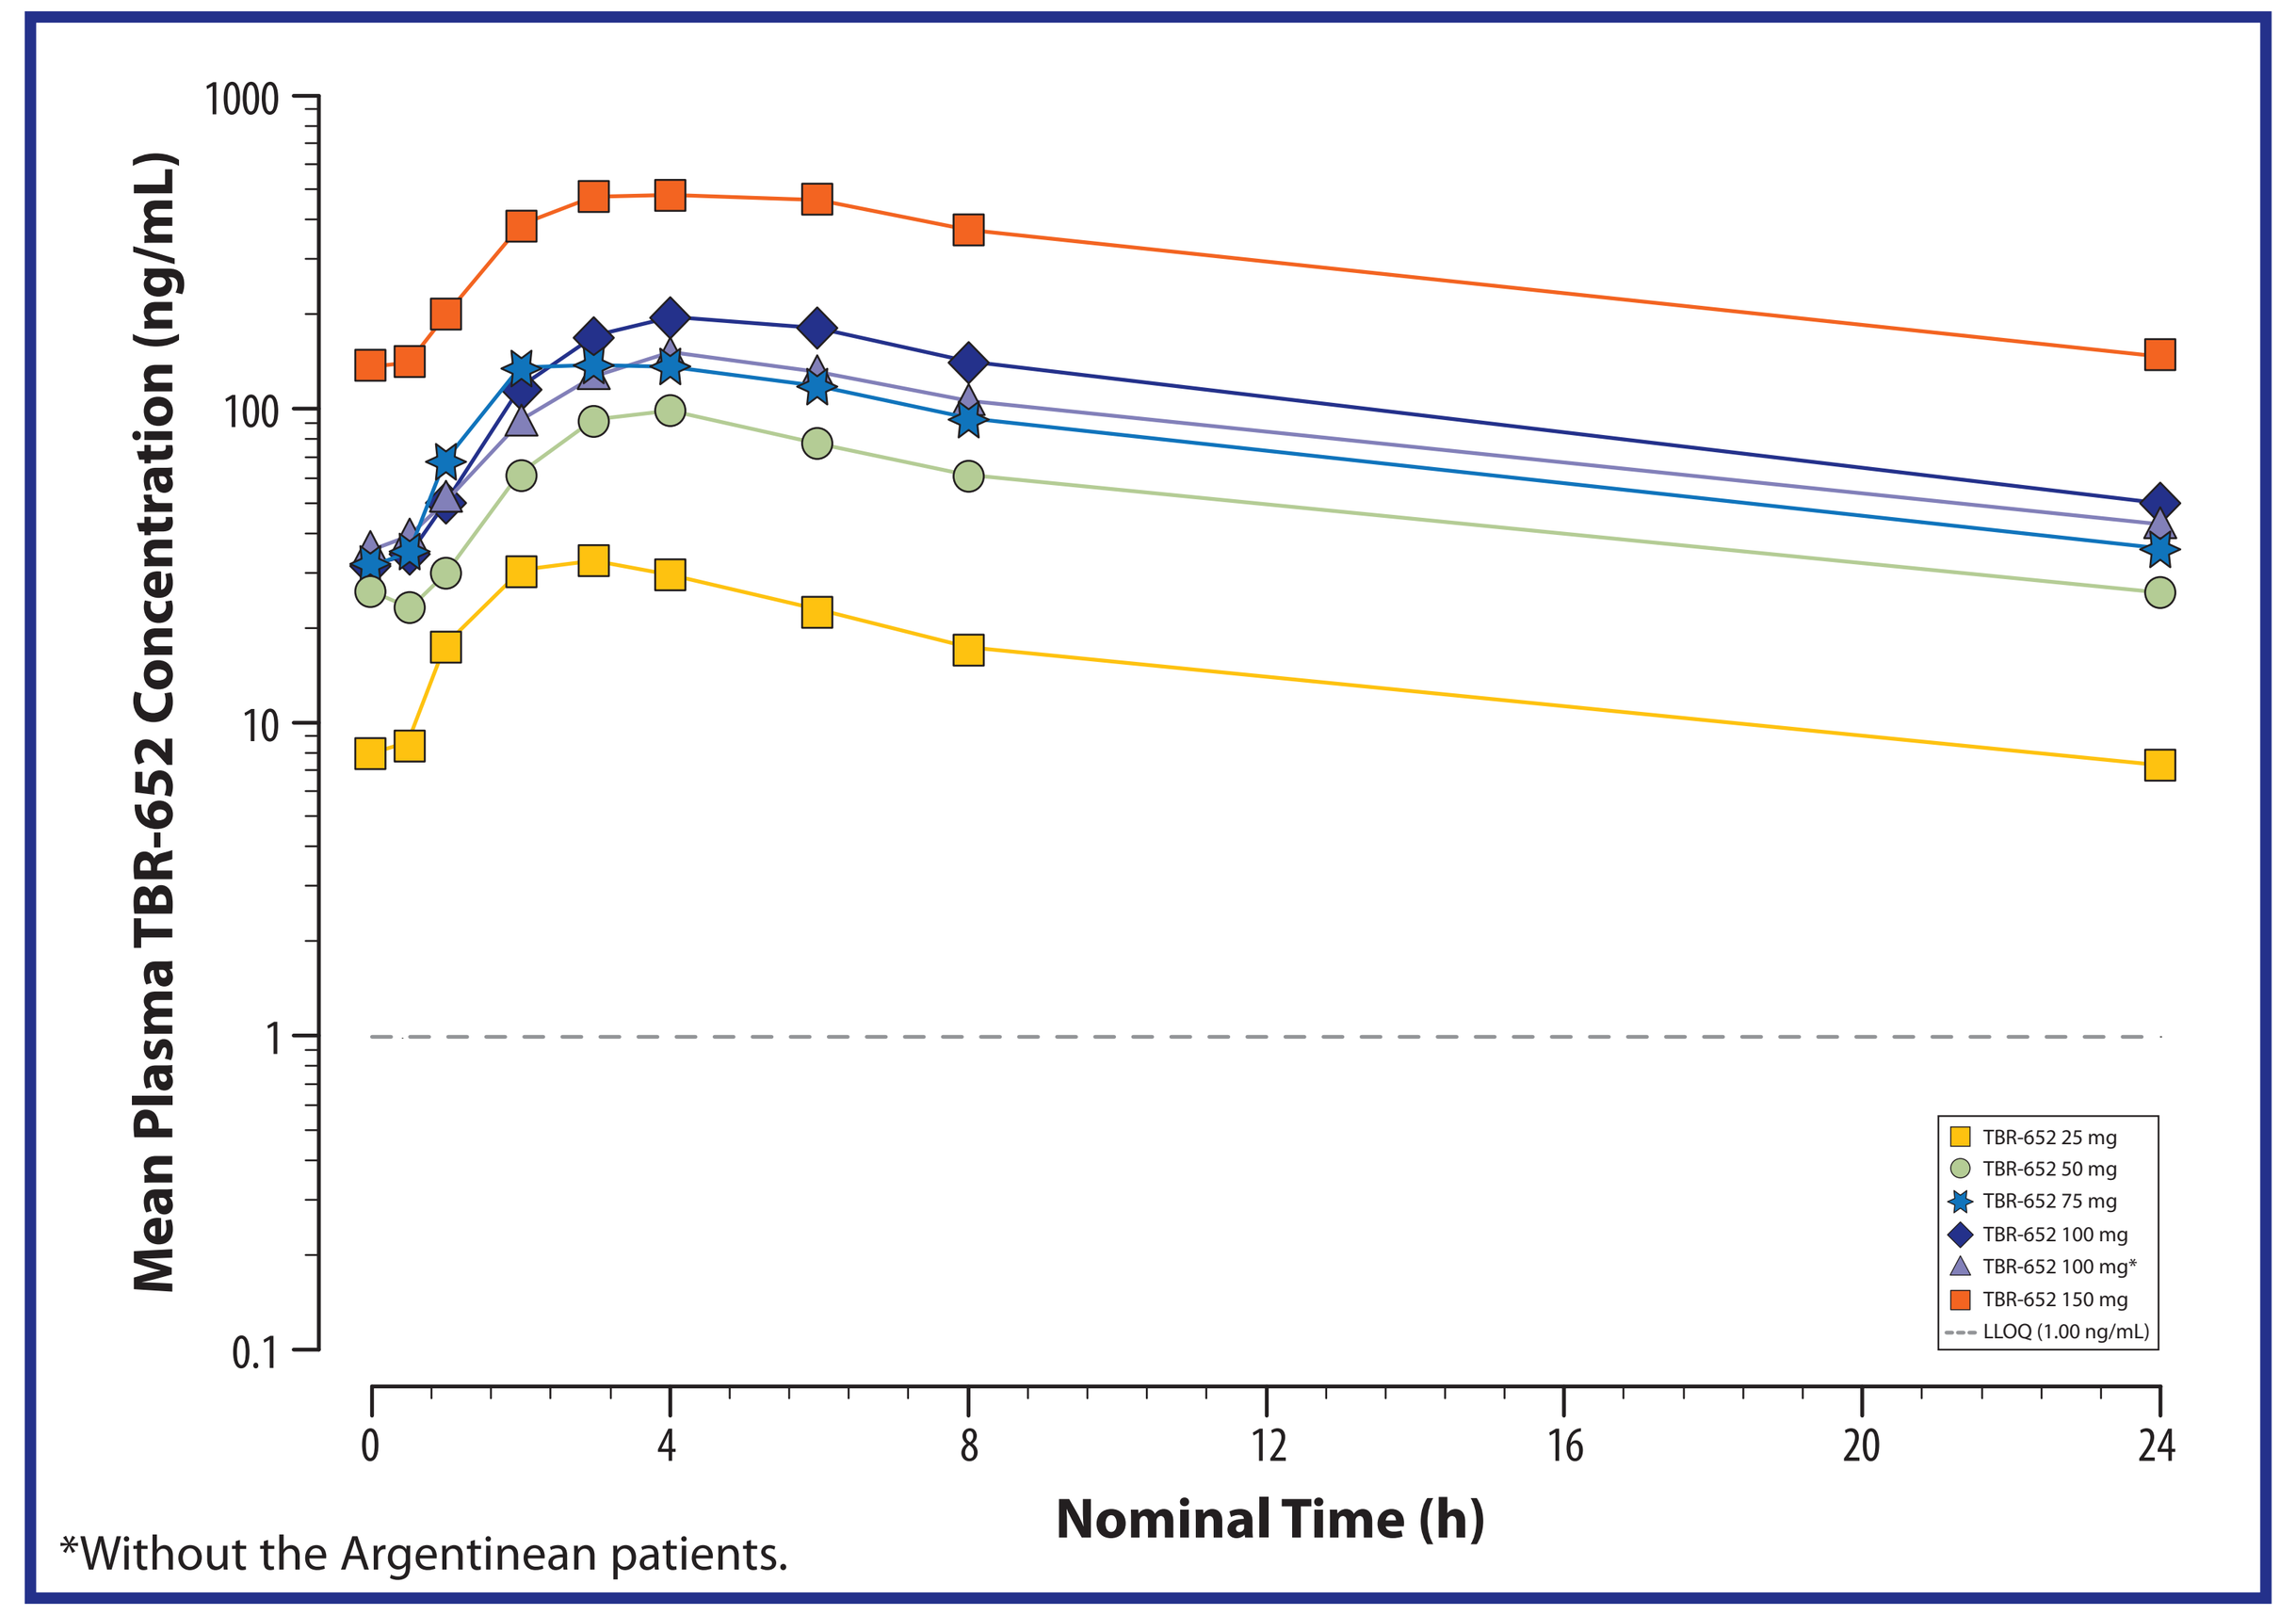
Mean Plasma TBR-652 Concentration (ng/mL)
Nominal Time (h)
TBR-652 25 mg
TBR-652 50 mg
TBR-652 75 mg
TBR-652 100 mg
TBR-652 100 mg*
TBR-652 150 mg
LLOQ (1.00 ng/mL)

*Without the Argentinean patients.

Figure 2. Median Plasma HIV-1 RNA-Time Profiles Following Oral QD TBR-652 Administrations for 10 Days and 30 Days of Follow-Up Period

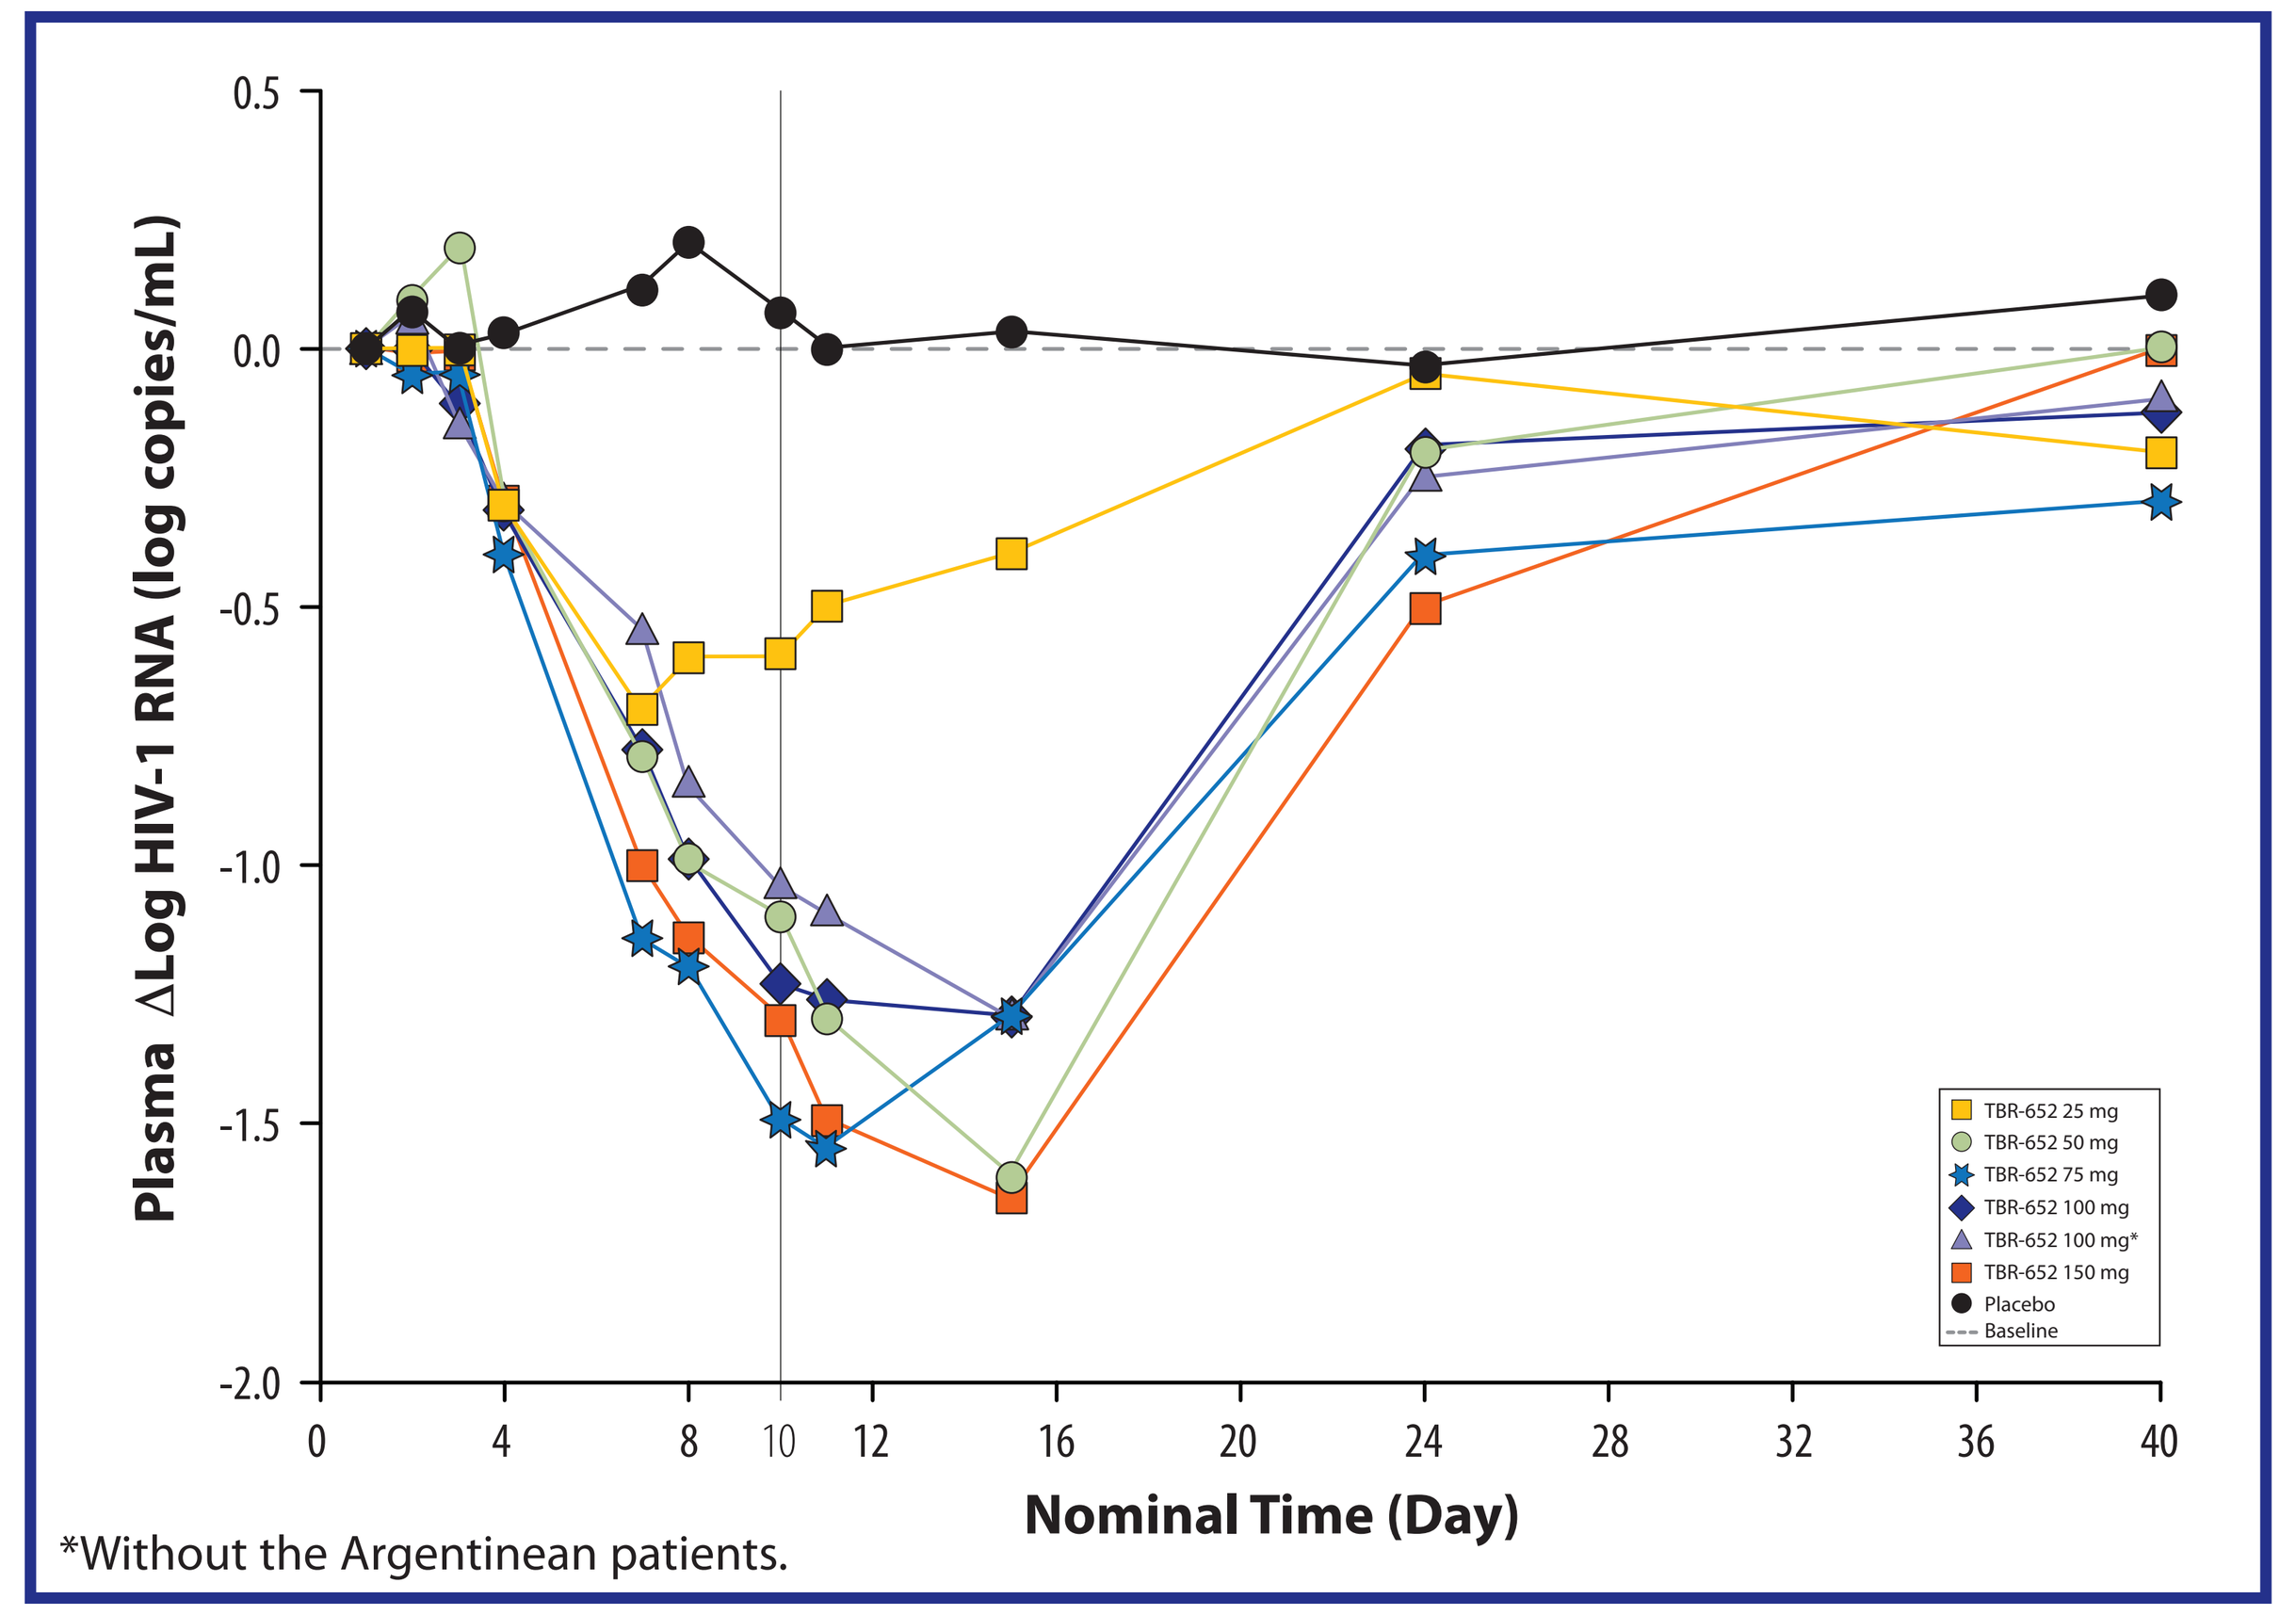
Plasma ΔLog HIV-1 RNA (log copies/mL)
Nominal Time (Day)
TBR-652 25 mg
TBR-652 50 mg
TBR-652 75 mg
TBR-652 100 mg
TBR-652 100 mg*
TBR-652 150 mg
Placebo
Baseline

*Without the Argentinean patients.

Table 1. Mean (CV%) PK and PD Parameters of TBR-652 in Plasma—Day 10

PK Parameters	Placebo	TBR-652 25 mg QD (n=9)	TBR-652 50 mg QD (n=7)	TBR-652 75 mg QD (n=8)	TBR-652 100 mg QD (n=10)	TBR-652 100 mg QD (n=8)[b]	TBR-652 150 mg QD (n=9)
AUC_{τ} (ng•h/mL)		382.0 (52.8)	1245 (40.4)	1916 (30.6)	2659 (52.8)	2088 (37.0)	7272 (36.8)
$AUC_{0\text{-last}}$ (ng•h/mL)		611.7 (70.3)	2650 (40.7)	3625 (29.9)	5068 (50.6)	4289 (48.1)	15390 (32.8)
C_{max} (ng/mL)		35.11 (46.9)	102.4 (41.9)	157.0 (41.0)	196.5 (54.3)	152.6 (35.5)	508.1 (34.9)
C_{min} (ng/mL)		6.88 (55.7)	21.5 (44.9)	30.1 (36.7)	30.0 (68.6)	32.5 (67.6)	128.0 (40.4)
C_{ss} (ng/mL)		15.9 (45.6)	48.9 (43.5)	79.8 (30.6)	111 (58.5)	87.0 (37.0)	317 (34.9)
T_{max}[a] (h)		2.98 (1.00 – 4.03)	4.00 (2.00 – 6.00)	3.50 (2.00 – 6.17)	4.00 (3.00 – 6.00)	3.92 (3.00 – 6.00)	4.00 (3.00 – 8.00)
$t_{½}$ (h)		22.50 (42.7)	47.62 (44.5)	29.78 (12.4)	32.06 (23.1)	33.04 (24.3)	41.26 (42.1)
CL_{ss}/F (L/h)		81.31 (45.3)	49.26 (55.3)	42.17 (27.8)	48.06 (51.2)	54.96 (40.9)	22.95 (32.3)
V_{ss}/F (L)		2106 (54.7)	2400 (69.5)	1424 (42.3)	1737 (67.7)	2044 (54.5)	910.3 (49.0)
MRT (h)		26.81 (34.3)	47.45 (37.2)	33.03 (18.8)	35.36 (35.7)	37.88 (33.8)	38.66 (22.6)
PD Parameters	**n=10**	**n=8[c]**	**n=7**	**n=8**	**n=10**	**n=8[b]**	**n=8[d]**
ΔLog HIV-1 RNA_{max} (log copies/mL)[a]	-0.3 (-0.8 – 0.1)	-0.7 (-2.0 – 0.0)	-1.7 (-2.2 – -0.8)	-1.8 (-2.1 – -1.0)	-1.4 (-2.2 – -0.9)	-1.3 (-2.5 – -0.9)	-1.7 (-2.2 – -0.7)
ΔLog HIV-1 RNA Day 11 (log copies/mL)[a]	0.1 (-0.5 – 0.3)	-0.5 (-1.8 – 0.4)	-1.3 (-1.3 – -0.5)	-1.6 (-1.9 – -1.0)	-1.2 (-1.8 – -0.8)	-1.1 (-1.8 – -0.8)	-1.5 (-2.0 – -0.7)

[a]Median (minimum–maximum).
[b]Without the Argentinean patients.
[c]The change from baseline could not be calculated for patient #1009 because no baseline value was available for this patient.
[d]One patient with dual/mixed tropic virus was excluded from the PD analyses.

PK Discussion

- Results of the Helmert Contrast confirm that steady state was reached by Day 8 of repeated once-daily dosing (*P*>0.05 for the comparison of predose on Day 8 vs mean predose on Days 9 and 10).
- Slope estimates describing the relationship between In-transformed PK parameters $AUC_{0\text{-}24}$ and C_{max} and In-transformed doses were greater than 1, suggesting that exposure levels of TBR-652 increased in more than a dose-proportional manner.
- Mean oral clearance of TBR-652 following repeated oral administrations of 25–150 mg TBR-652 decreased with increasing doses (from 81.3 L/h to 23.0 L/h for the 25- to 150-mg QD dose levels, respectively). Consistent with the slightly lower CL/F reported for the higher dose levels, mean half-life of TBR-652 also increased with increasing doses.
- Two patients who were dosed at the Argentina study site (IDs 3076 and 3077) and who were randomized to the 100-mg QD treatment group displayed increased exposure of TBR-652 with levels as high as 2-fold the mean exposure levels observed for the 100-mg dose level. This difference may be related to an altered exposure to TBR-652 resulting from coadministration with a meal of higher-fat content as compared to the patients dosed in the US clinics. Descriptive statistics for the 100-mg dose group were computed with and without these 2 patients. Modest differences in mean PK profiles and parameters were observed.

PD Discussion

- Median log_{10} viral load decline from baseline increased with increasing dose levels on Day 11. The median ΔLog HIV-1 RNA for the 25 mg, 50 mg, 75 mg, 100 mg, and 150 mg TBR-652 dose levels were -0.5, -1.3, -1.6, -1.2, and -1.5 log_{10} copies/mL, respectively.
- Median maximum viral load decline ranged from -0.7 to -1.8 log copies/mL on Day 11. The one-sided ANOVA comparison of change from baseline viral load between treated and placebo groups showed a significant difference in viral load suppression between all treated groups versus placebo from study Day 4 through Day 15.

PD/PK Discussion

Table 2. Model Selection—AIC Values for the Different PK/PD Models Tested

Model	AUC_{τ}	C_{max}	C_{ss}
Simple E_{max}	66.8	66.9	66.8
E_{max} Sigmoid	68.2	68.5	68.2
Simple E_{max} (In-transformed)	70.1	68.9	68.7
Sigmoid E_{max} (In-transformed)	68.4	68.8	68.6
Linear	75.2	74.3	75.2

A simple E_{max} model was found to best describe the relationship between PK and PD of TBR-652 (no sigmoid function) based on the minimization of the AIC. Results obtained from the simple E_{max} model are presented in **Table 2** and a figure depicting the E_{max} model is presented in **Figure 3**.

Figure 3. Correlation Between C_{ss} (ng/mL) vs ΔLog HIV-1 RNA (log copies/mL) on Day 11

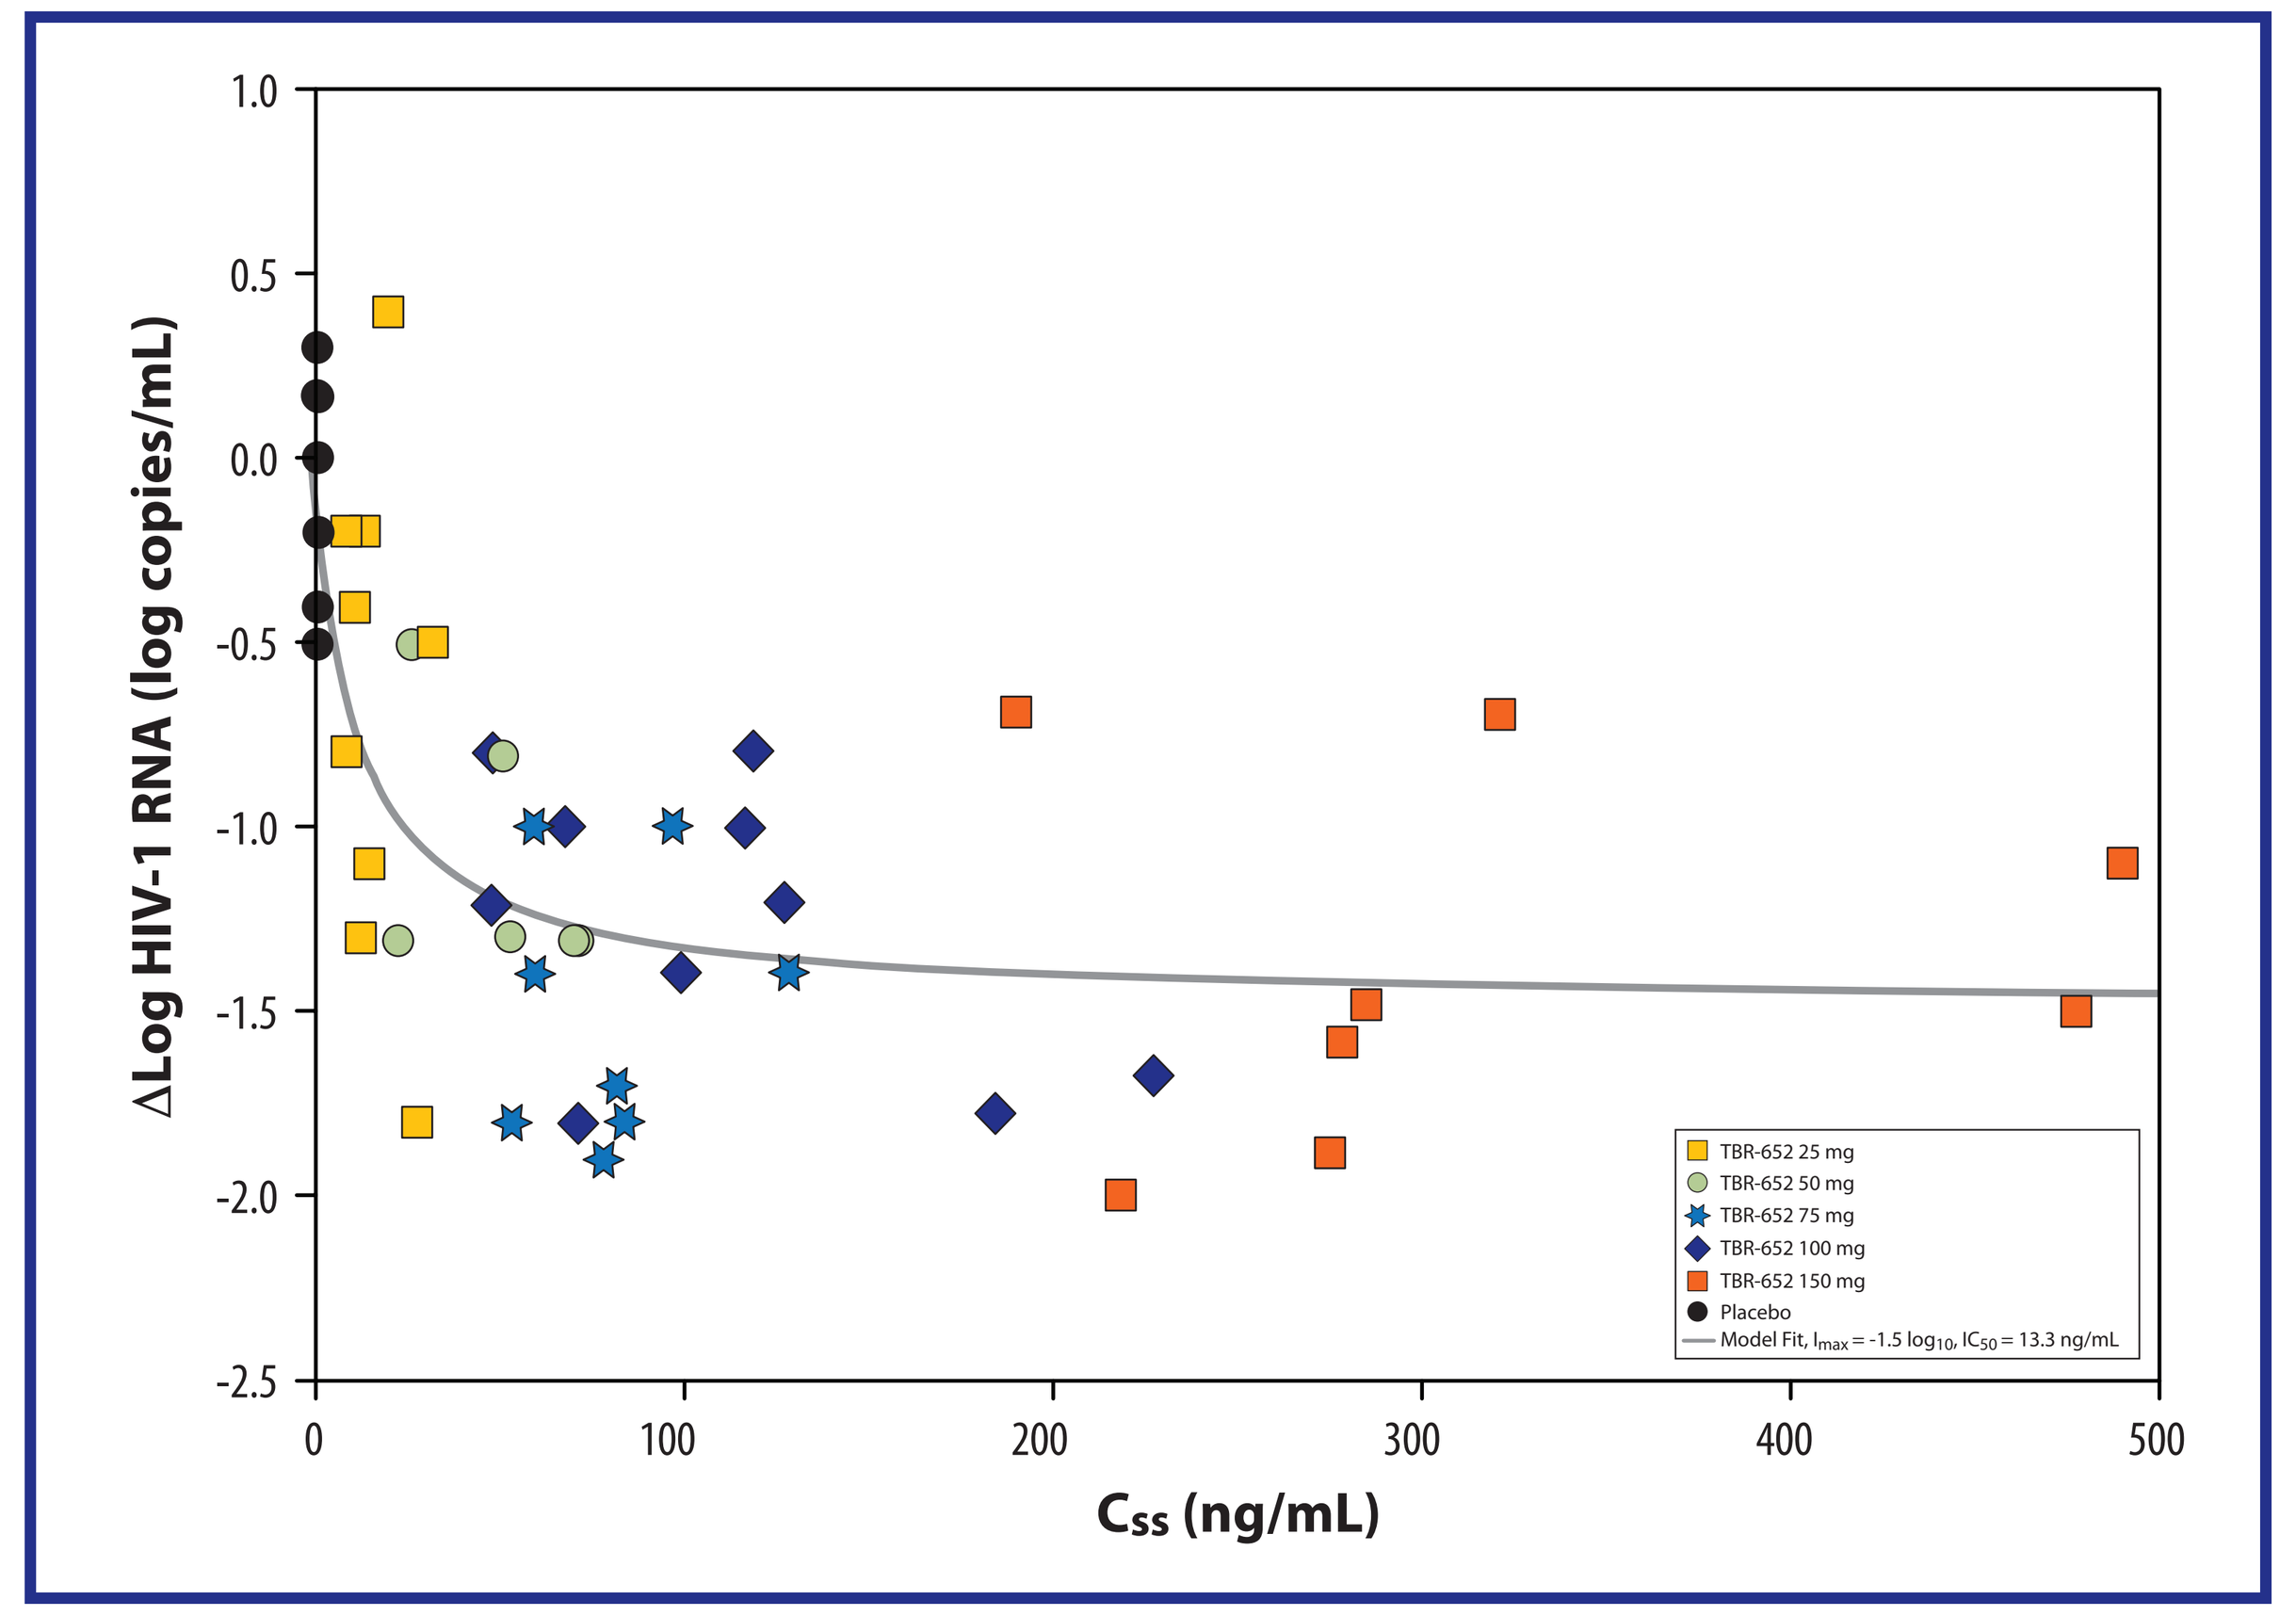
ΔLog HIV-1 RNA (log copies/mL)
Css (ng/mL)
TBR-652 25 mg
TBR-652 50 mg
TBR-652 75 mg
TBR-652 100 mg
TBR-652 150 mg
Placebo
Model Fit, Imax = -1.5 log10, IC50 = 13.3 ng/mL

CONCLUSIONS

- Ten days of treatment with TBR-652 was generally well tolerated with no dose-limiting adverse events identified.
- Treatment with TBR-652 resulted in potent, dose-dependent decreases in viral load with median nadir decreases up to 1.8 log_{10} copies/mL with significant viral load suppression persisting during the 30-day observation period.
- The relationship between TBR-652 exposure and antiviral activity was described by an inhibitory E_{max} model.
- Steady-state plasma TBR-652 concentrations of 13.3 ng/mL are expected to provide 50% of the maximal inhibition. The maximum antiviral effect of TBR-652 (I_{max}) determined with the PK/PD model was -1.5 log_{10} copies/mL.
- The data from this study support the further development of TBR-652 in HIV-infected patients. Plans for Phase 2b clinical development are underway.

REFERENCE

1. Smith BP, Vandenhende FR, DeSanta KA, et al. Confidence interval criteria for assessment of dose proportionality. *Pharm Res*. 2000;17(10):1278–1283.

ACKNOWLEDGEMENTS

Thanks to the investigators, the patients, and all the study personnel.

The 17th Conference on Retroviruses and Opportunistic Infections
February 16–19, 2010; San Francisco, California